A Unique CRISPR-Based Nuclease with a Non-NGG PAM Efficiently Targets Multiple Exclusive Genomic Sites for Immuno-Oncology Based Therapy

Bar Ben Baruch, Yonit Ben David, Sigal Cohen, Maya Noff, Nir Shahar, Ira Gotliv, Rafi Emmanuel, Lior Izhar

Emendo Biotherapeutics

emendo bio
NEXT GENERATION CRISPR
MAKING ANY GENE TARGETABLE

INTRODUCTION

Chimeric antigen receptor (CAR)-T cell therapy is a recent clinically successful approach to tackling cancer, in which T cells are genetically modified to allow specific recognition and efficient killing of cancer cells via tumor associated antigens[1]. Current CAR-based treatments require costly and time-consuming autologous cell transfer. Patients' own cells are frequently of low quality and difficult to obtain, further supporting the advantage of an allogeneic transplantation. To prevent graft versus host reactions and avoid host-mediated rejection of healthy donor-derived allogeneic cells, these cells must be adjusted by eliminating the expression of endogenous recognition components, such as T cell receptor alpha constant (TRAC or TCR) and CD3, and of HLA class I molecules such as β-2 microglobulin (B2M)[2,3]. In this study, we tested the feasibility of gene editing in allogeneic cells using our novel high-fidelity CRISPR-associated nuclease OMNIA4. OMNIA4 is a highly active nuclease with a unique non-NGG PAM recognition domain. The unique PAM allows gene-editing in exclusive genomic sequences that are not accessible by commonly used nucleases. Our strategy involves disrupting endogenous T cell recognition elements as well as checkpoint receptors and exhaustion genes that restrict anti-tumor T cell response. RNA guides (gRNAs) were designed for several of such genes of interest and their editing via OMNIA4 nuclease was evaluated by NGS in HeLa cells. Ribonucleoproteins (RNPs), including OMNIA4 and gRNAs designed to target distinctive sites in either TRAC, CD3e or B2M genes, were applied to primary T cells. The resulting editing outcome was evaluated by measuring T cell receptor (TCR), CD3e or B2M surface expression by flow cytometry. In addition, an unbiased analysis was performed to identify 'off-targets' edited by the nuclease. Our gRNA screening yielded at least one (and up to five) active gRNA for each gene of interest, with editing level >70%. Flow cytometric analysis showed that editing in primary T cells resulted in about 94% TCR negative cells, about 85% CD3e negative cells and about 95% B2M negative cells. An unbiased assay revealed no off-targets for neither of the guides. These findings demonstrate efficient, accurate and safe impairment of a self-presenting element and endogenous T cell recognition components. Our approach offers gene editing at unique targets as a tool to generate universal allogeneic T cells that could be employed in the development of 'off-the-shelf' 'ready-to-use' CAR-T therapeutic agents for large-scale clinical applications.

METHODS

T cells from one donor were thawed and activated by T Cell Activation/ Expansion Kit (cat:130-091-441) according to manufacturer instructions. Then, a cleavage activity assay was done with OMNIA4 + sgRNA. After 7 days cells were analyzed by flow cytometry and the rest of the samples were used for editing analysis by NGS.

OMNIA4

OMNIA4	
Protein Length (AA)	1349 (161.9 KDa)
gRNA length (nt)	107+22=129
PAM (TXTL results)	NNRACT

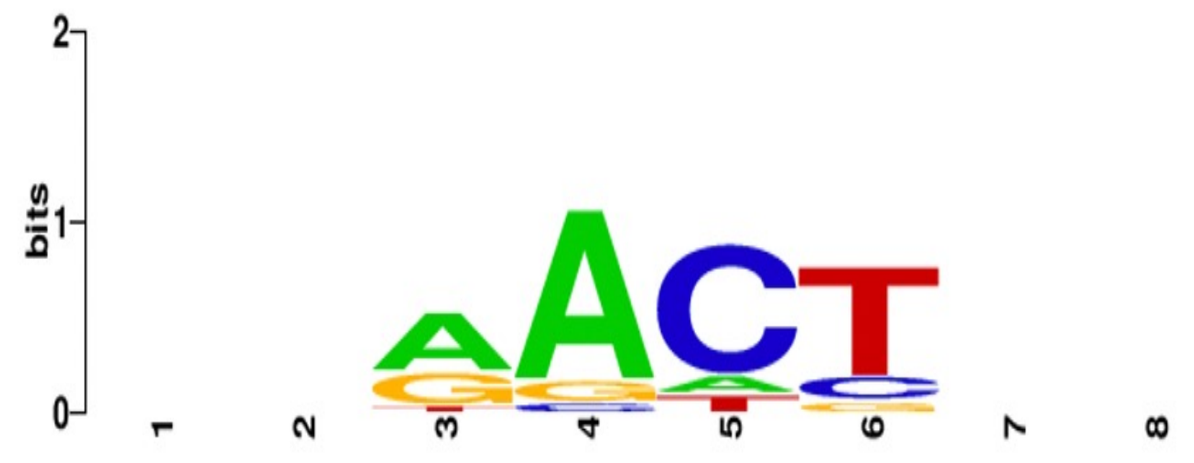
bits
2
1
0
1
2
3
4
5
6
7
8

	# of patent families directed to guide sequences	# of NGG guides left after patent screen	# of guides for non-NGG nuclease OMNIA4
TRAC	56	0/20	15
TRBC1	23	1/20	3
TRBC2	23	1/21	5
CD3e	4	10/79	18
B2M	42	0/33	12
CIITA	9	0/294	35
PD1 (PDCD1)	51	0/196	29
TET2	6	8/638	288

- Commonly used nucleases (NGG PAM) and compatible guide targets are heavily patented by multiple unrelated entities.
- An unmet need for a nuclease with a unique non-NGG PAM recognition domain.
- The unique PAM allows gene-editing in exclusive genomic targets.

RESULTS

B2M

- B2M is a component of MHC class I molecules
- KO of B2M aims at preventing an immune response from cytotoxic CD8+ T cells

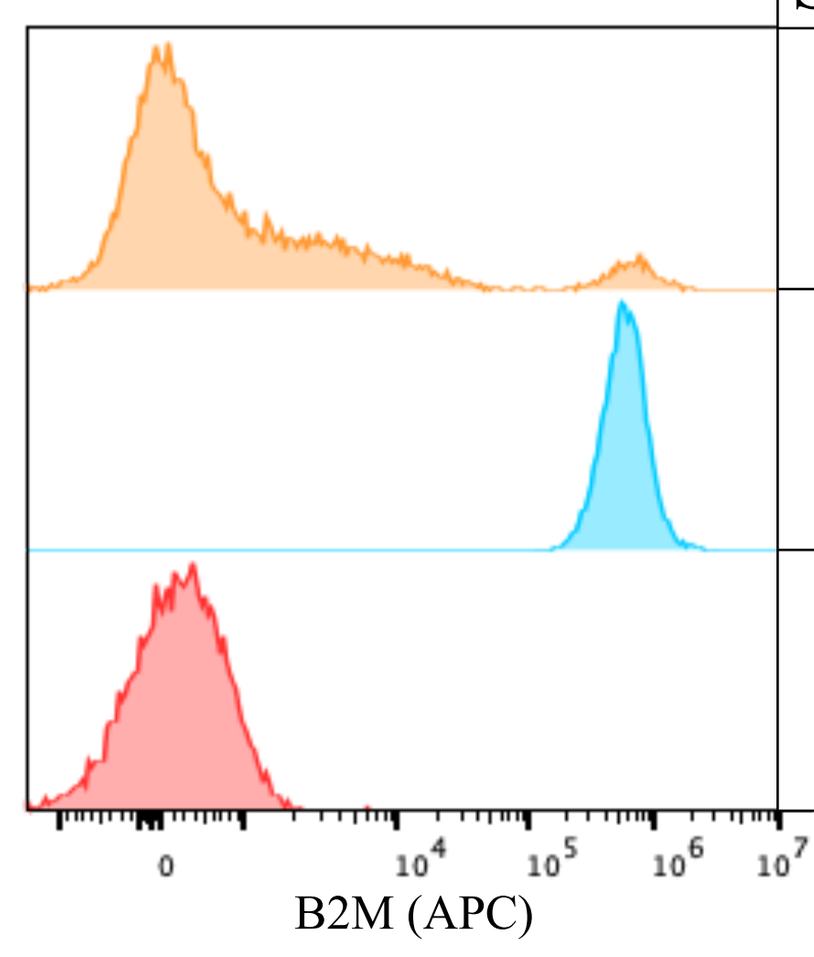
0
10^4 10^5 10^6 10^7
B2M (APC)

Sample Name	% Editing FACS	GM	% Editing NGS
Edited	94.7	921	55.2
NT	0.45	56200	0.01
REA	99.9	245	

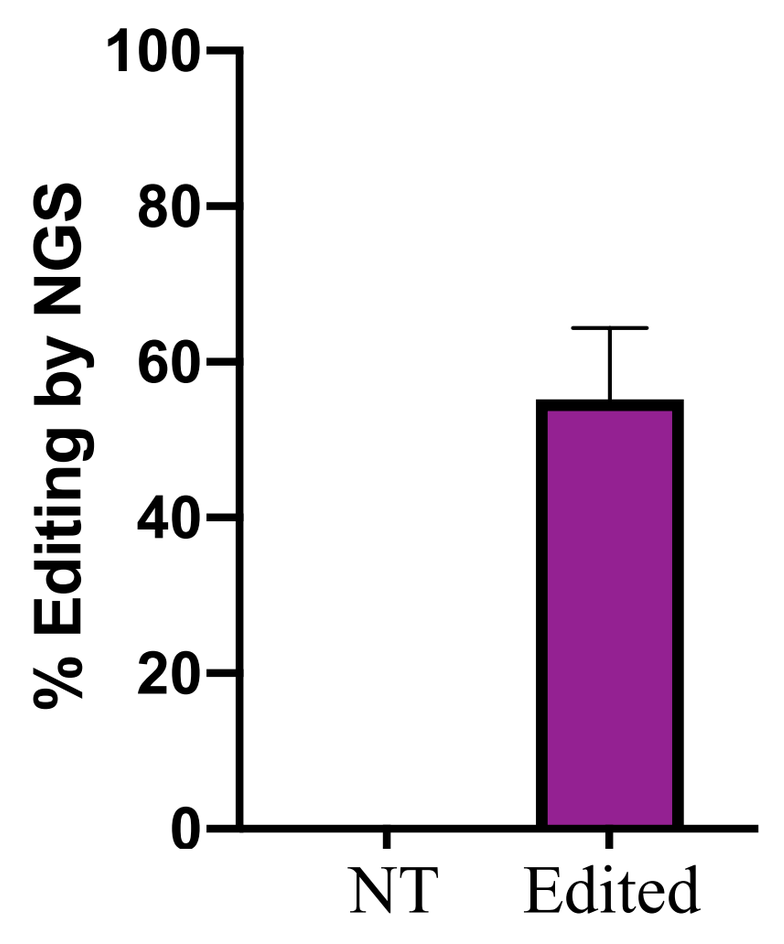
% Editing by NGS
100
80
60
40
20
0
NT
Edited

B2M_KO: 95% editing (FACS) and 55% editing (NGS) after 7 days

TRAC

- TRAC is a constant region of the T cell receptor (TR) alpha chain
- TRAC KO aims at eliminating graft-versus-host disease (GVHD)

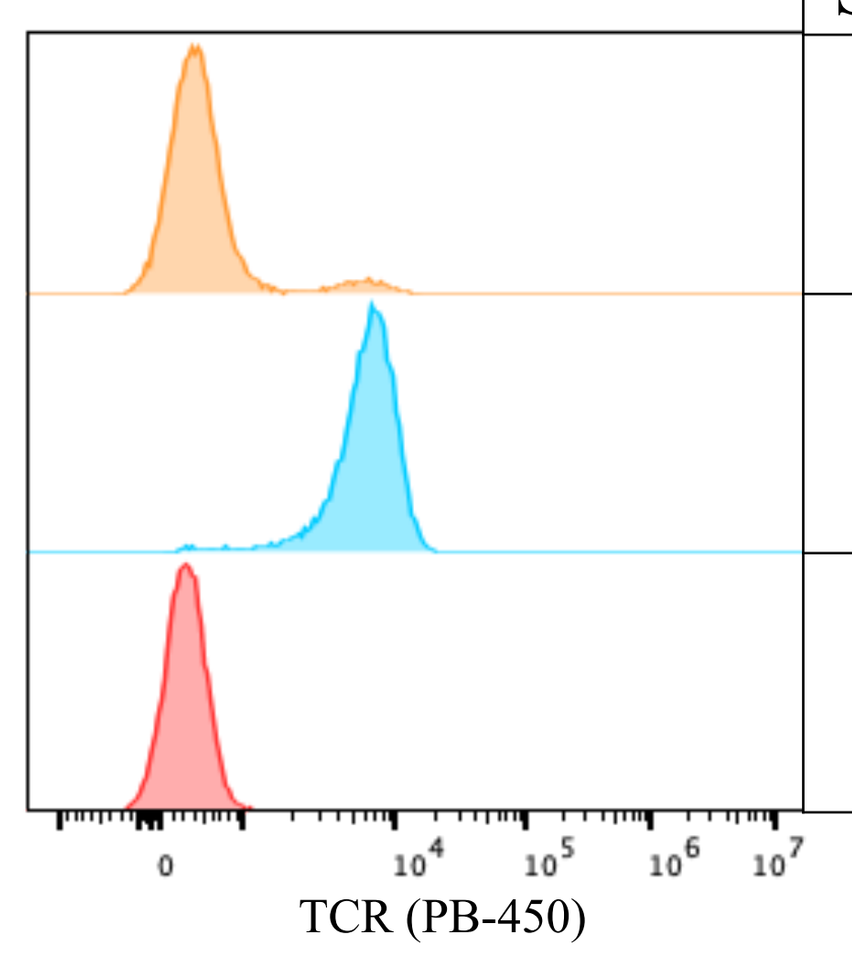
0
10^4 10^5 10^6 10^7
TCR (PB-450)

Sample Name	% Editing FACS	GM	% Editing NGS
Edited	94.3	529	89.6
NT	5.44	5832	0.02
REA	100	330	

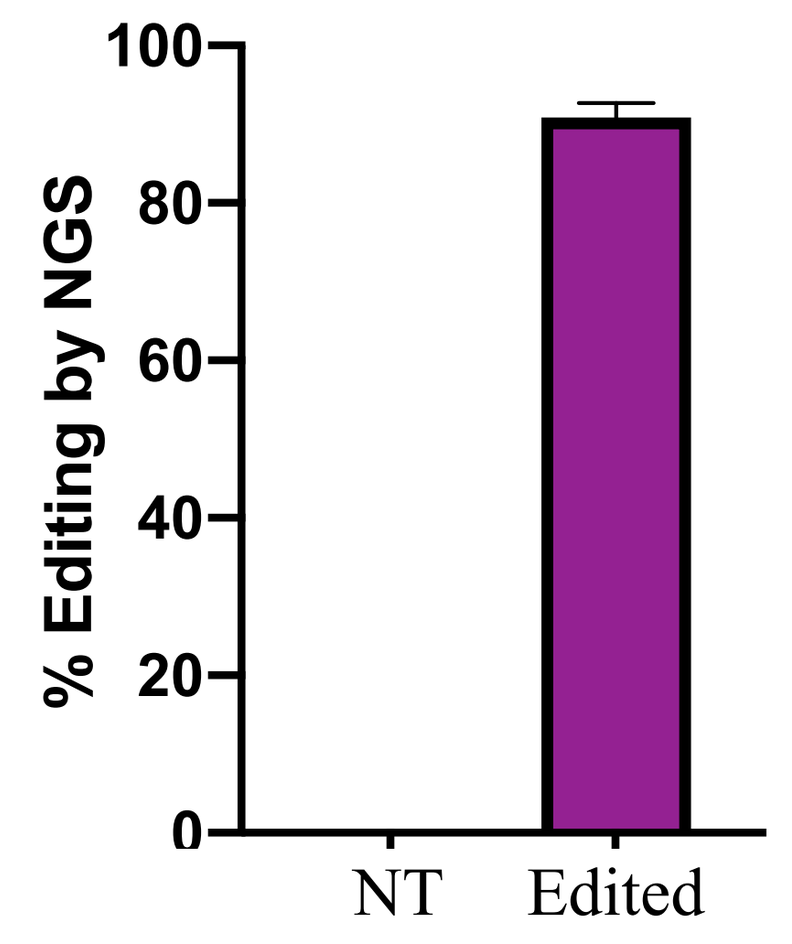
% Editing by NGS
100
80
60
40
20
0
NT
Edited

TRAC_KO: 94% editing (FACS) and 90% editing (NGS) after 7 days

CD3e

- CD3e codes to a polypeptide, which is part of the T-cell receptor-CD3 complex
- This complex plays an important role in coupling antigen recognition to intracellular signal-transduction pathways

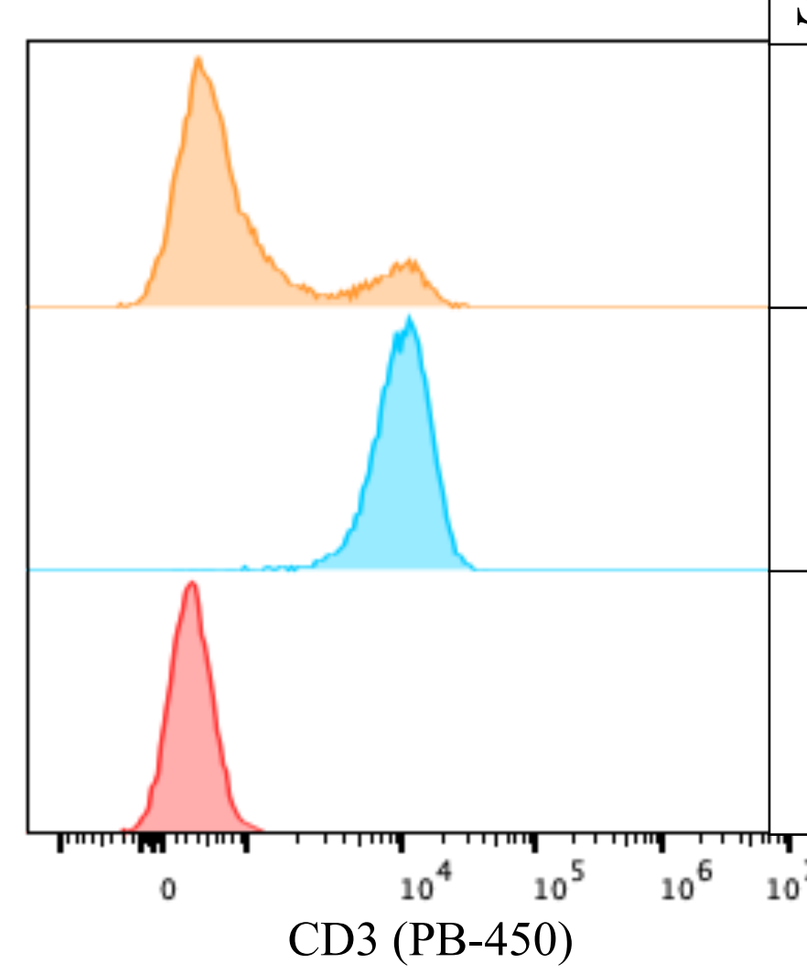
0
10^4 10^5 10^6 10^7
CD3 (PB-450)

Sample Name	% Editing FACS	GM	% Editing NGS
Edited	84.5	912	70.3
NT	1.41	9449	0.03
REA	100	354	

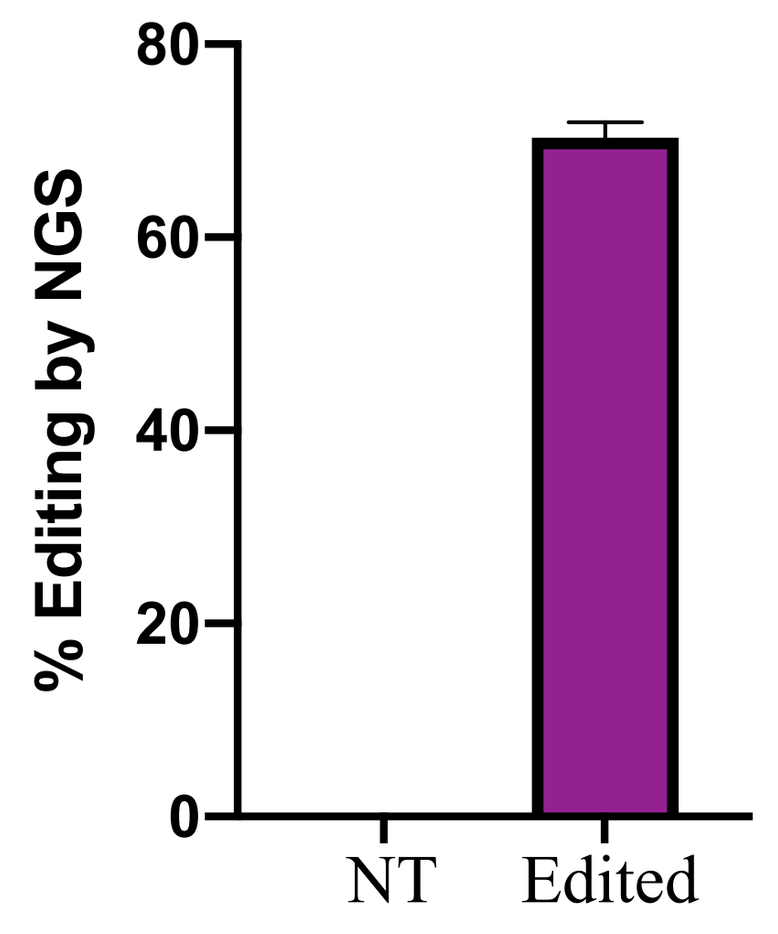
% Editing by NGS
80
60
40
20
0
NT
Edited

CD3e_KO: 85% editing (FACS) and 70% editing (NGS) after 7 days

TIM3

- TIM3 expression, especially in conjunction with PDCD1 (PD-1), induces an exhausted T cell state

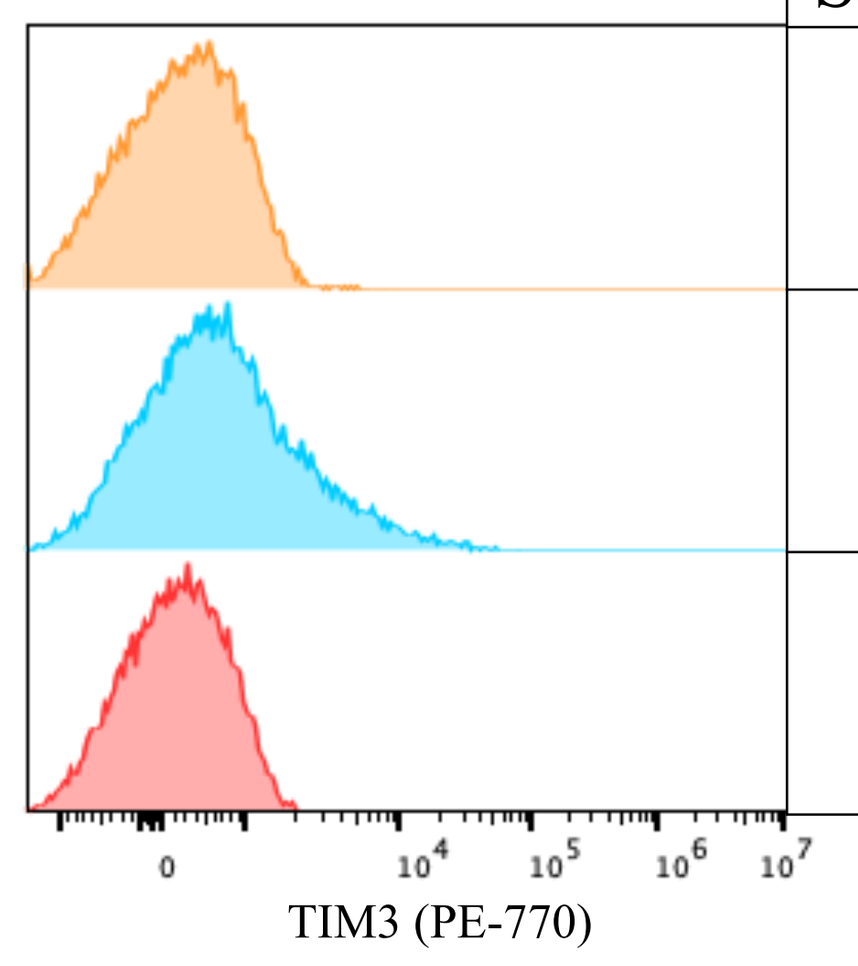
0
10^4 10^5 10^6 10^7
TIM3 (PE-770)

Sample Name	% TIM3 Positive	GM	% Editing NGS
Edited	2.02	258	93.6
NT	19.6	689	0.04
REA	0.43	201	

* Editing analysis by FACS is presented after normalization to NT cells.

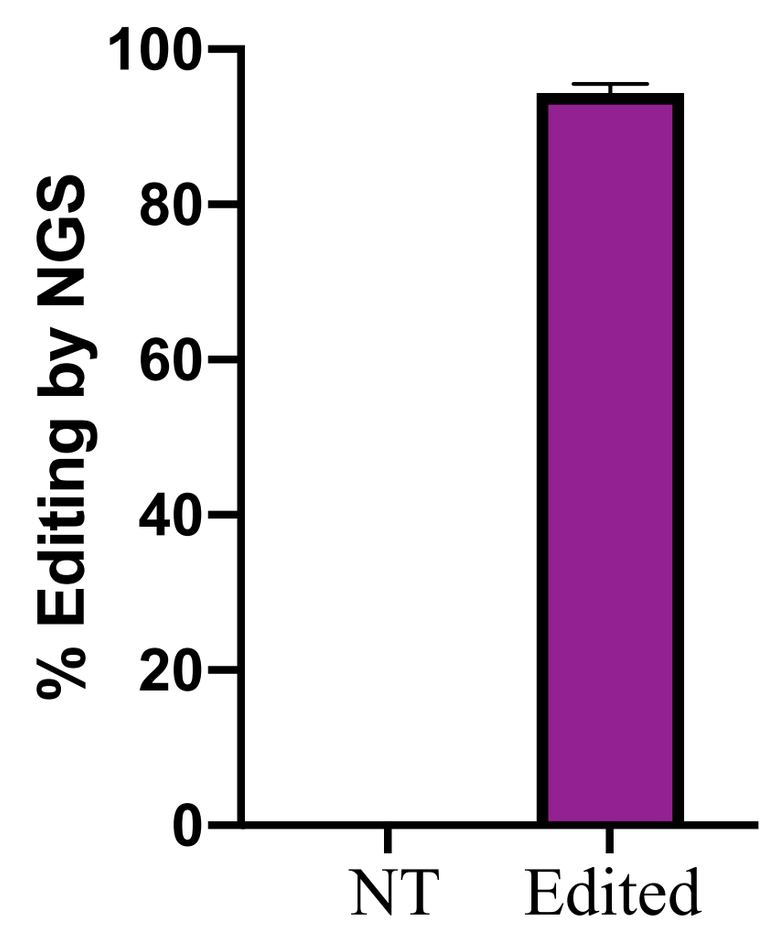
% Editing by NGS
100
80
60
40
20
0
NT
Edited

TIM3_KO: 90% editing (FACS)* and 94% editing (NGS) after 7 days

CONCLUSIONS

Allogeneic CAR T Cell Production Using OMNIA4

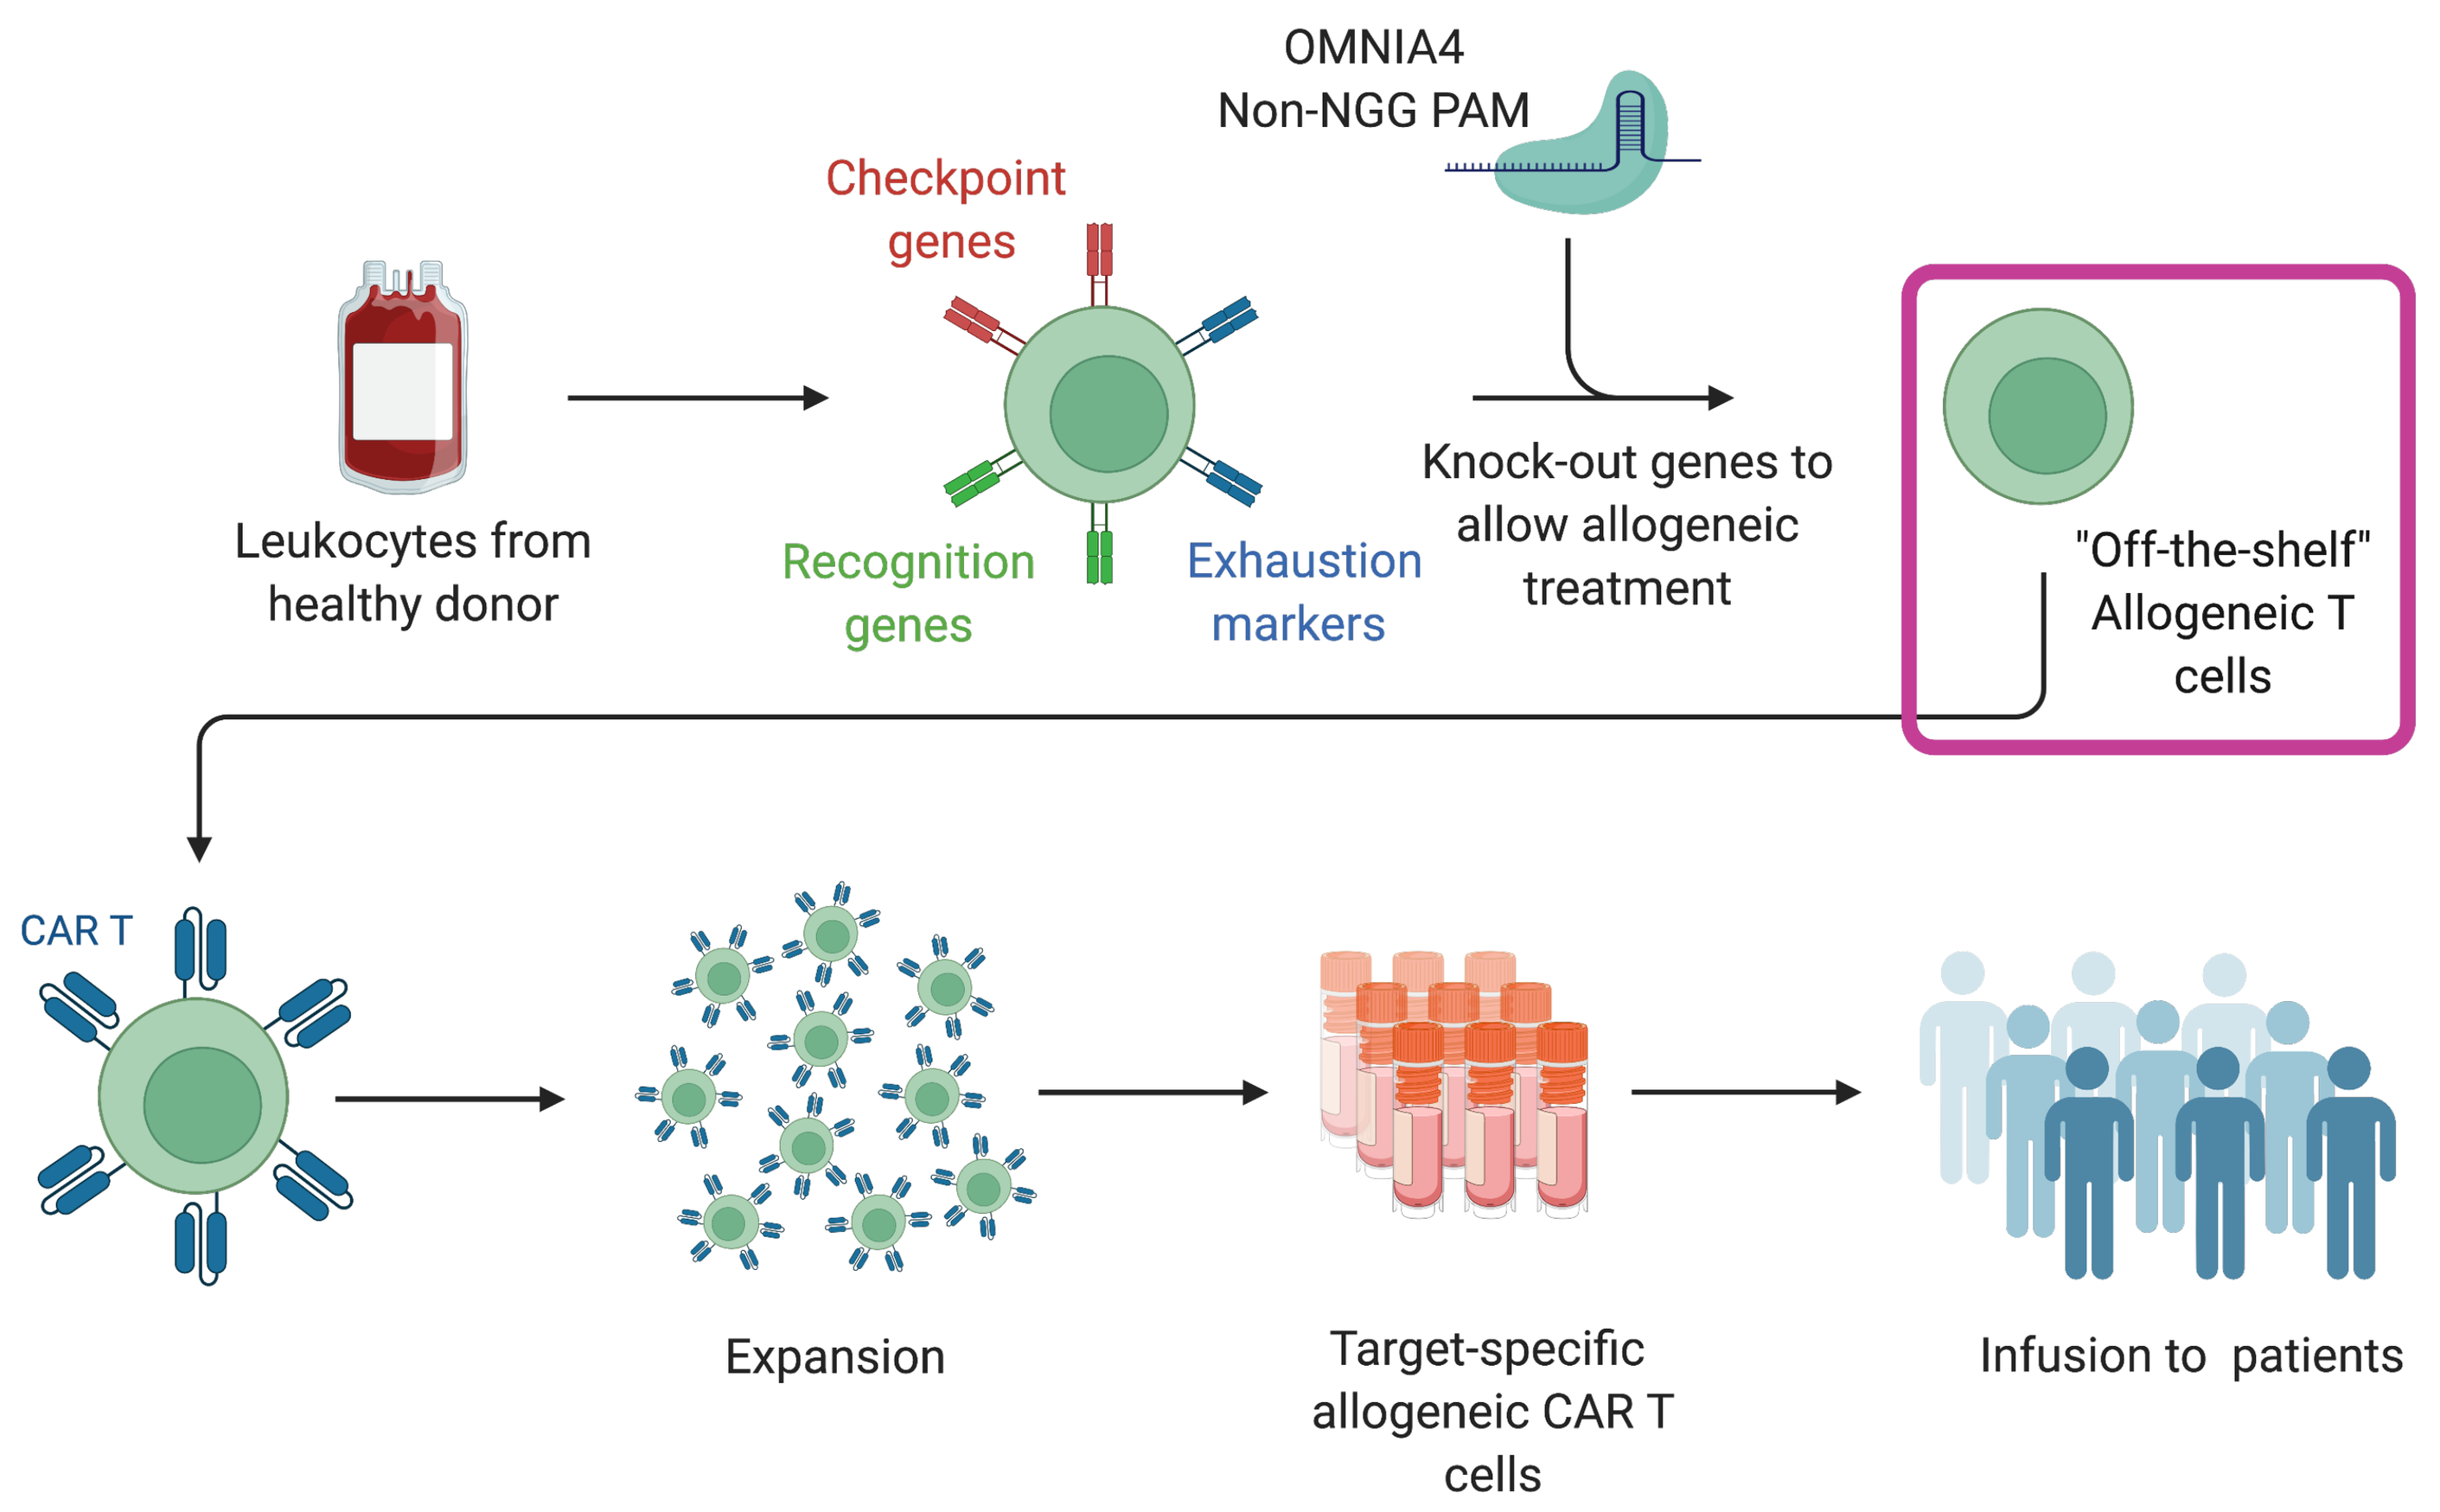
OMNIA4
Non-NGG PAM
Checkpoint genes
Leukocytes from healthy donor
Recognition genes
Exhaustion markers
Knock-out genes to allow allogeneic treatment
"Off-the-shelf" Allogeneic T cells
CAR T
Expansion
Target-specific allogeneic CAR T cells
Infusion to patients

- **Our approach offers gene editing at unique targets as a tool to generate universal allogeneic T cells that could be employed in the development of 'off-the-shelf' 'ready-to-use' CAR-T therapeutic agents for large-scale clinical applications.**
- **Whole package: Nuclease, sgRNA, PAM and a working protocol.**

REFERENCES

1. Labanieh, L., Majzner, R.G., and Mackall, C.L. (2018). Programming CAR-T cells to kill cancer. Nat. Biomed. Eng. *2*, 377–391.

2. Liu, X., Zhang, Y., Cheng, C., Cheng, A.W., Zhang, X., Li, N., Xia, C., Wei, X., Liu, X., and Wang, H. (2017). CRISPR-Cas9-mediated multiplex gene editing in CAR-T cells. Cell Res. *27*, 154–157.

3. Depil, S., Duchateau, P., Grupp, S.A., Mufti, G., and Poirot, L. (2020). 'Off-the-shelf' allogeneic CAR T cells: development and challenges. Nat. Rev. Drug Discov. *19*, 185–199.

RESEARCH POSTER PRESENTATION DESIGN © 2019
www.PosterPresentations.com