A Panel of Novel Engineered Nucleases Unlocks the Full Potential of Genome Editing

Noa Wertheimer, Nir Shpak, Noa Oded-Elkayam, Moran Azaria, Ella Segal

EmendoBio, Inc., New York, USA

ABSTRACT

A successful CRISPR based gene editing strategy requires developing an optimized composition of nuclease and gRNA per target gene per organ. Different editing strategies require different gene editing tools with specific nuclease characteristics, such as nuclease size, diverse PAM recognition, compatibility with delivery systems and high activity and specificity to the target site. A panel of nucleases, the OMNI nuclease series, was developed by EmendoBio to overcome such key challenges in the field. The OMNI nucleases recognize different PAM sequences that cover approx. 80% of the human genome; they vary in size; and they are compatible with all common delivery modalities, including AAV. OMNI nucleases are engineered to be as active as industry gold standard nucleases while displaying enhanced specificity, which eliminates off-target effects and enables allele specific editing. EmendoBio's technology creates tailor-made nucleases by combining a discovery pipeline and cutting-edge protein-engineering capabilities, which is supported by extensive computational tools, including proprietary AI.

The specificity and efficiency of the OMNI nucleases enabled an achievement of high HDR efficiency in multiple target cells including HSCs and more, and recently OMNI editors were development to achieve efficient DNA writing efficiency.

EmendoBio's panel of novel nucleases, and its proprietary techniques to customize such nucleases, enables expansion of the existing CRISPR toolbox and overcomes major gene editing challenges, offering solutions for a wide range of diseases.

OMNI discovery pipeline

Mining metagenomic data → **Classification into Types** → **Filter by Similarity to known nucleases** → **sgRNA prediction from CRISPR locus** → **OMNI candidates list+related guide**

Nuclease Portfolio

- 10,000 discovered nucleases
- 300 validated in vitro
- 80 shown active in cells
- 12 characterized
- 3 engineered

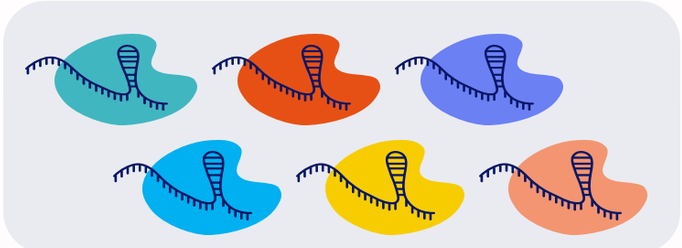

OMNI™ Genomic PAM Coverage

The diversity of PAM sites of the OMNI™ nucleases overcomes PAM constraints and significantly widens genome accessibility – making **any gene targetable**

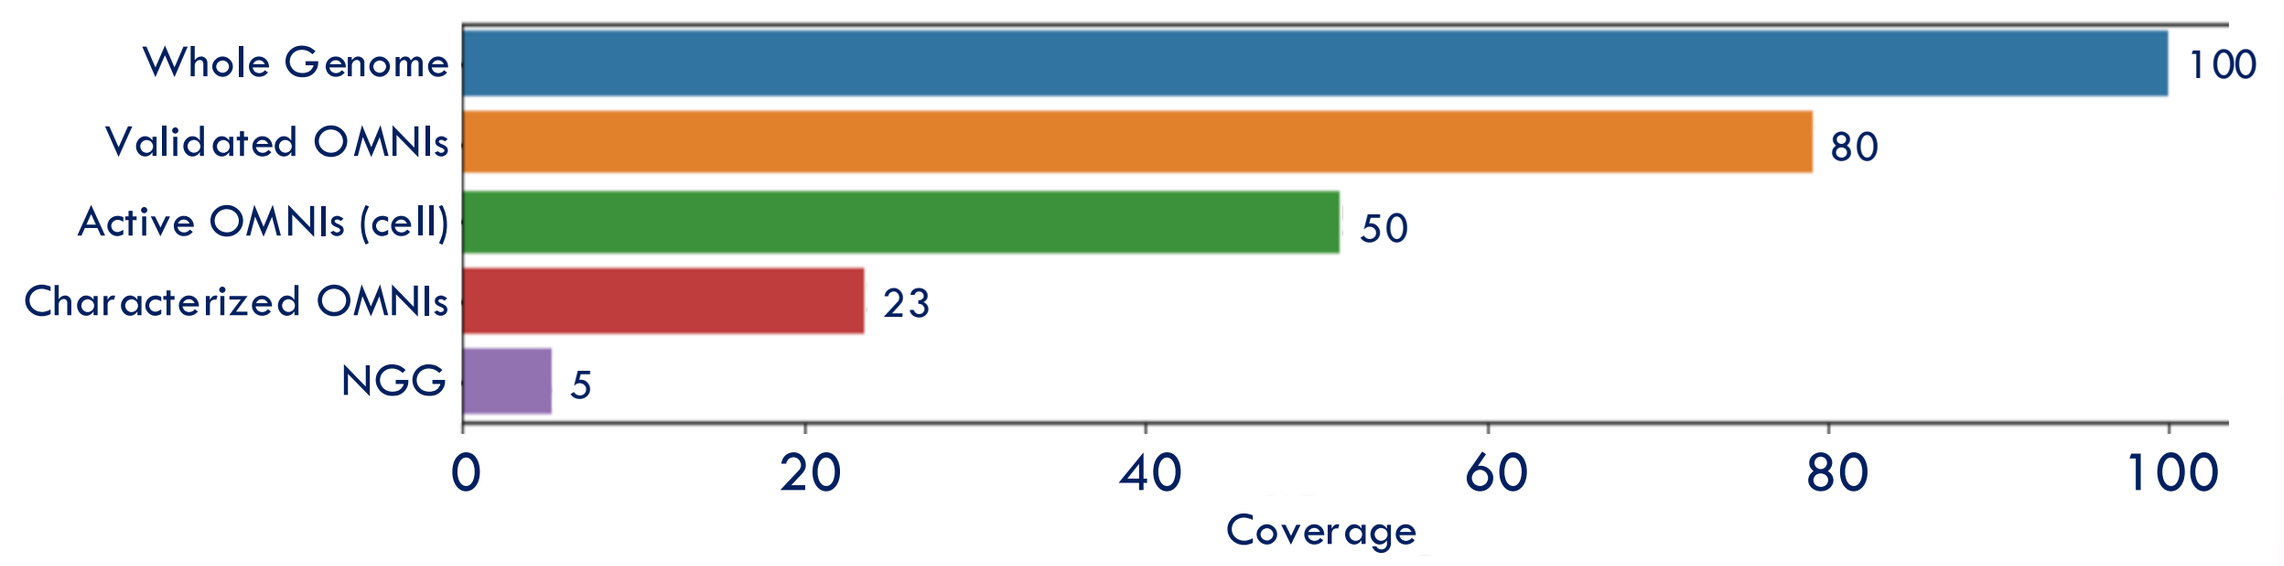

Whole Genome 100
Validated OMNIs 80
Active OMNIs (cell) 50
Characterized OMNIs 23
NGG 5
0 20 40 60 80 100
Coverage

OMNI engineering pipeline

Leading OMNIs → **Variants engineering** → **Variants productions** → **In-Vitro characterization** → **Cell-based validations** → **Novel OMNI** (• Highly active • Highly specific)

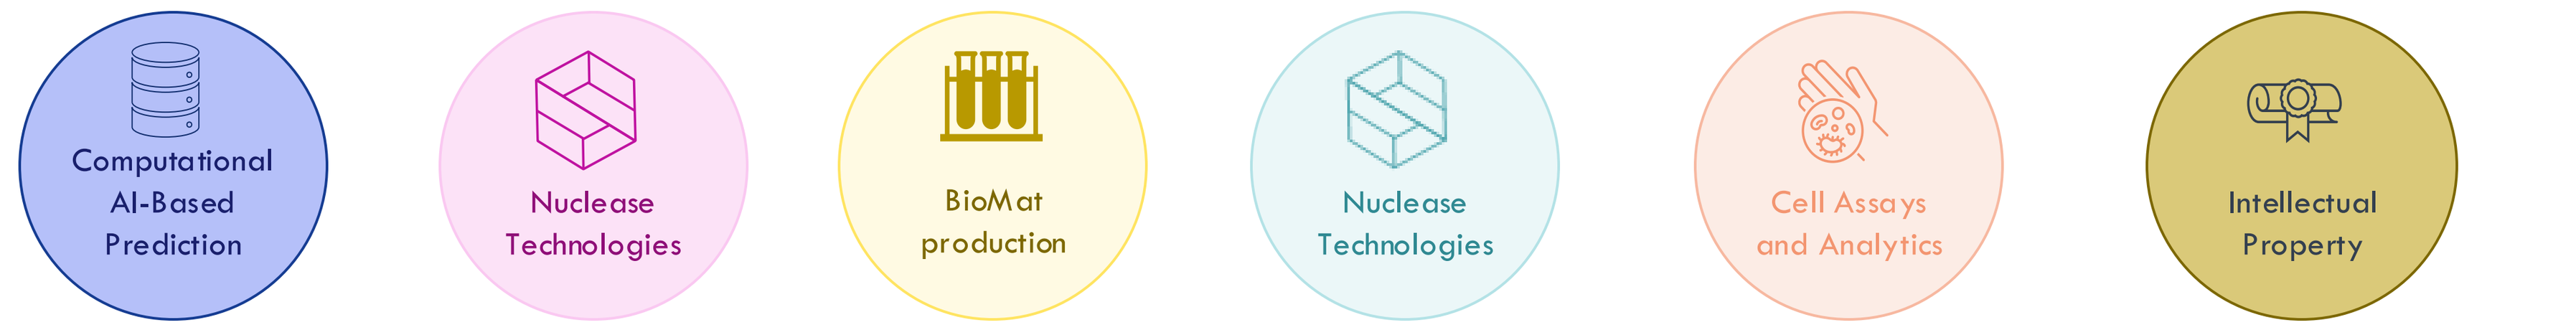

Computational AI-Based Prediction
Nuclease Technologies
BioMat production
Nuclease Technologies
Cell Assays and Analytics
Intellectual Property

Engineered OMNI variants demonstrate improved capabilities compared to non-engineered nucleases

The OMNI Platform - combining discovery and engineering for the generation of a broad portfolio of nucleases

1. OMNI-A1 - Increased specificity

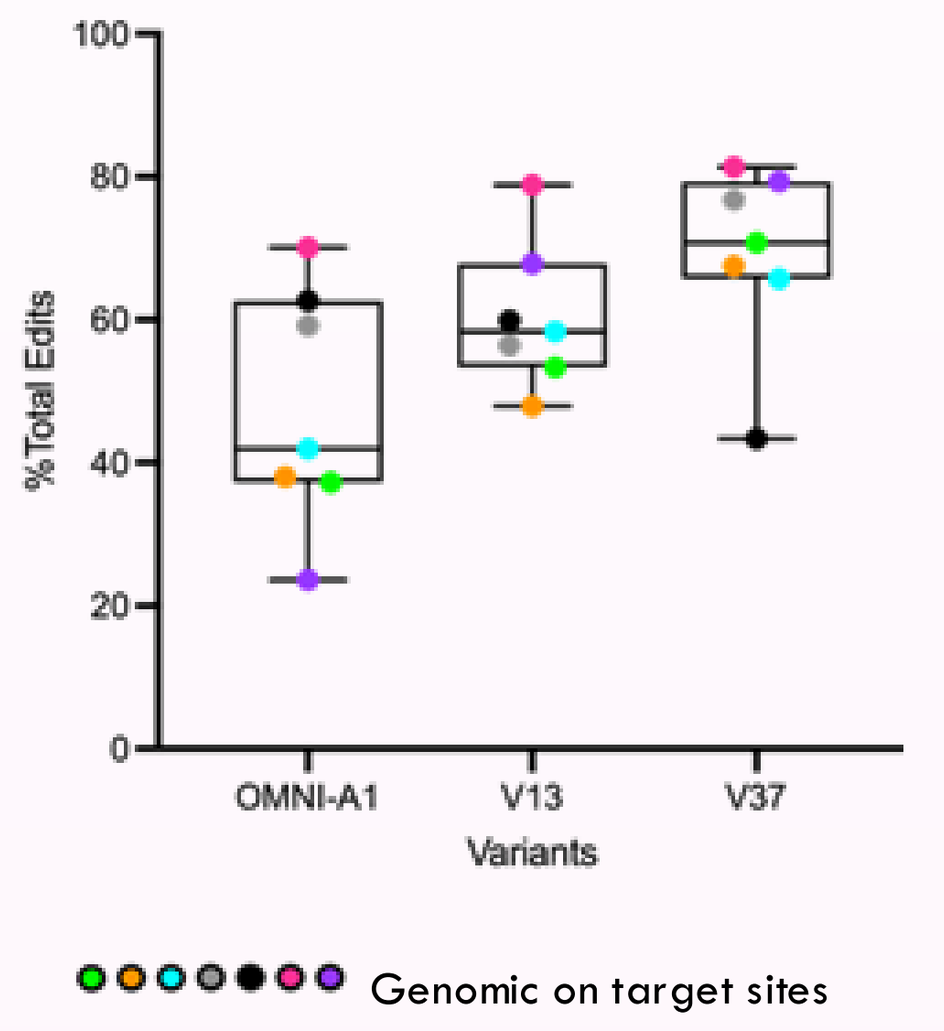

On targets
%Total Edits
OMNI-A1 V13 V37
Variants
Genomic on target sites

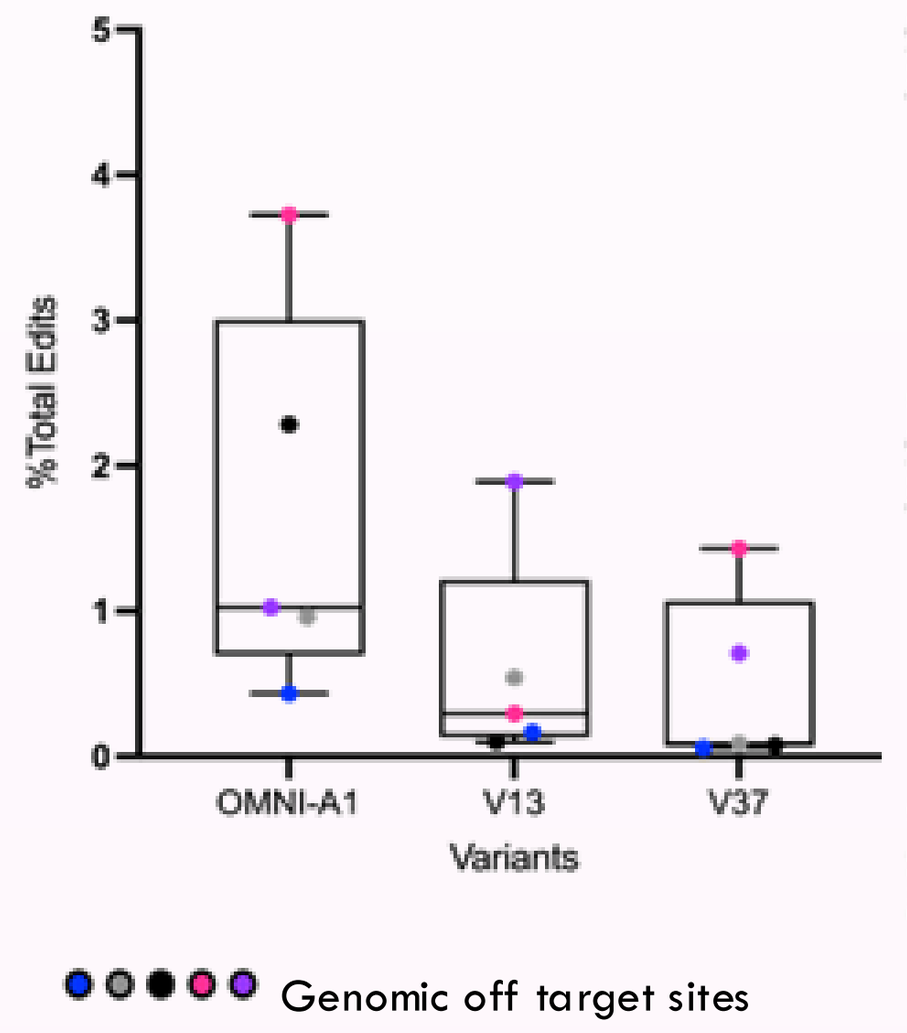

Off targets
%Total Edits
OMNI-A1 V13 V37
Variants
Genomic off target sites

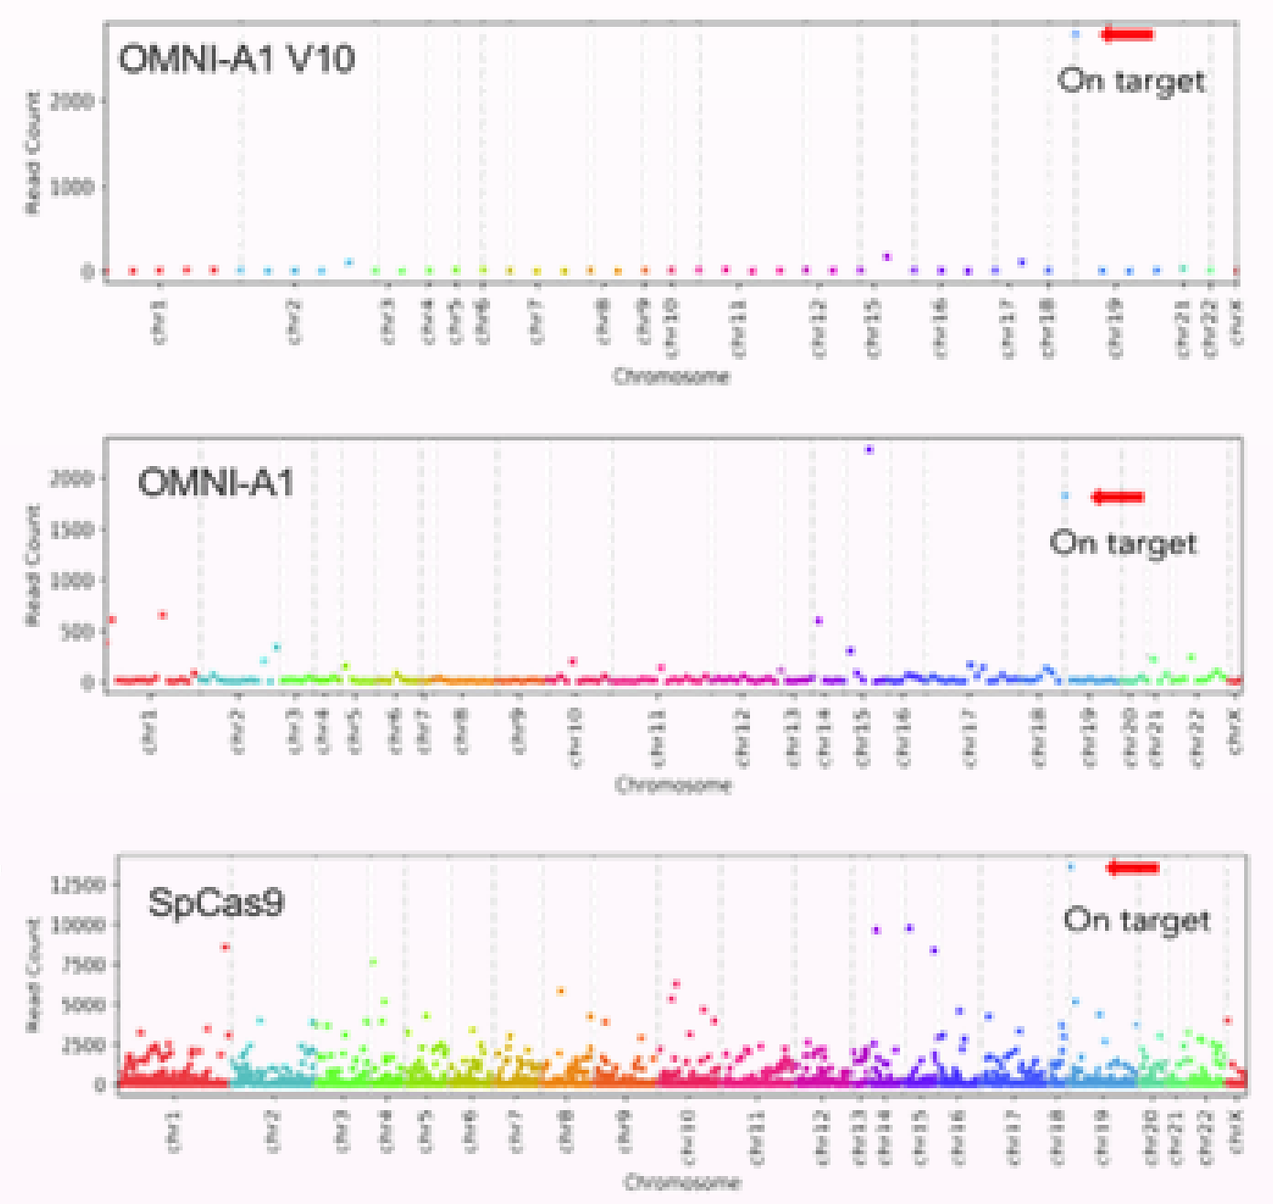

Unbiased Off-target Analysis
(in-vitro)
OMNI-A1 V10
On target
OMNI-A1
On target
SpCas9
On target
Chromosome

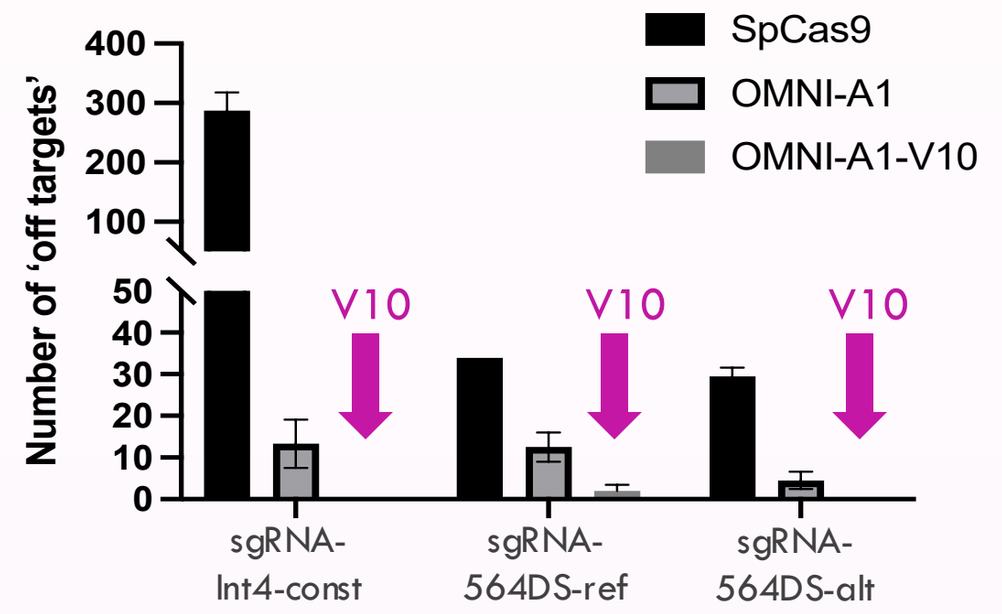

Unbiased Off-target Analysis
(cell based)
SpCas9
OMNI-A1
OMNI-A1-V10
Number of 'off targets'
V10
V10
V10
sgRNA-Int4-const
sgRNA-564DS-ref
sgRNA-564DS-alt

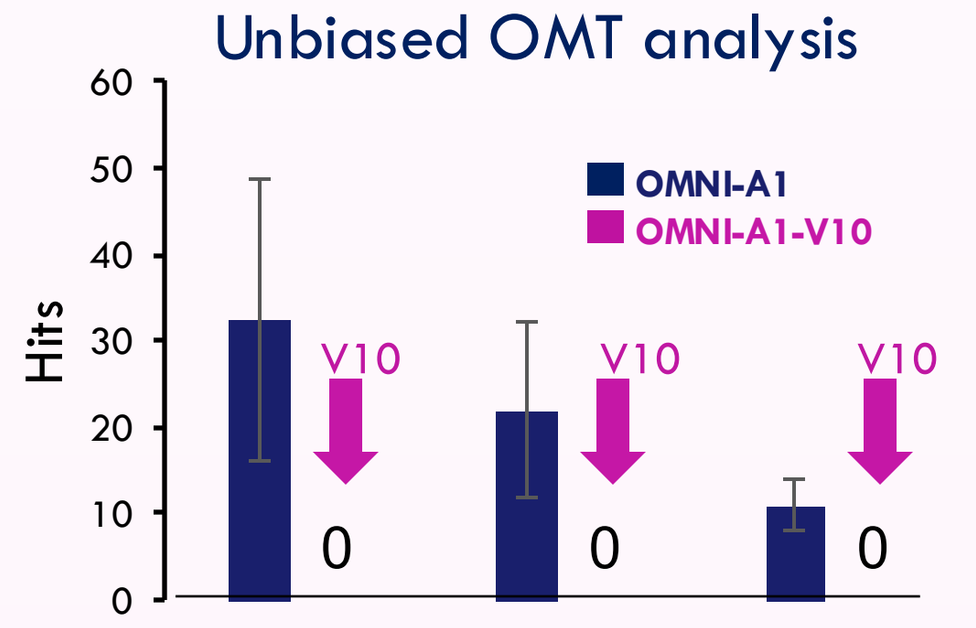

Unbiased OMT analysis
OMNI-A1
OMNI-A1-V10
Hits
V10
V10
V10
0
0
0

2. OMNI-A2 - AAV compatible nuclease with increased activity

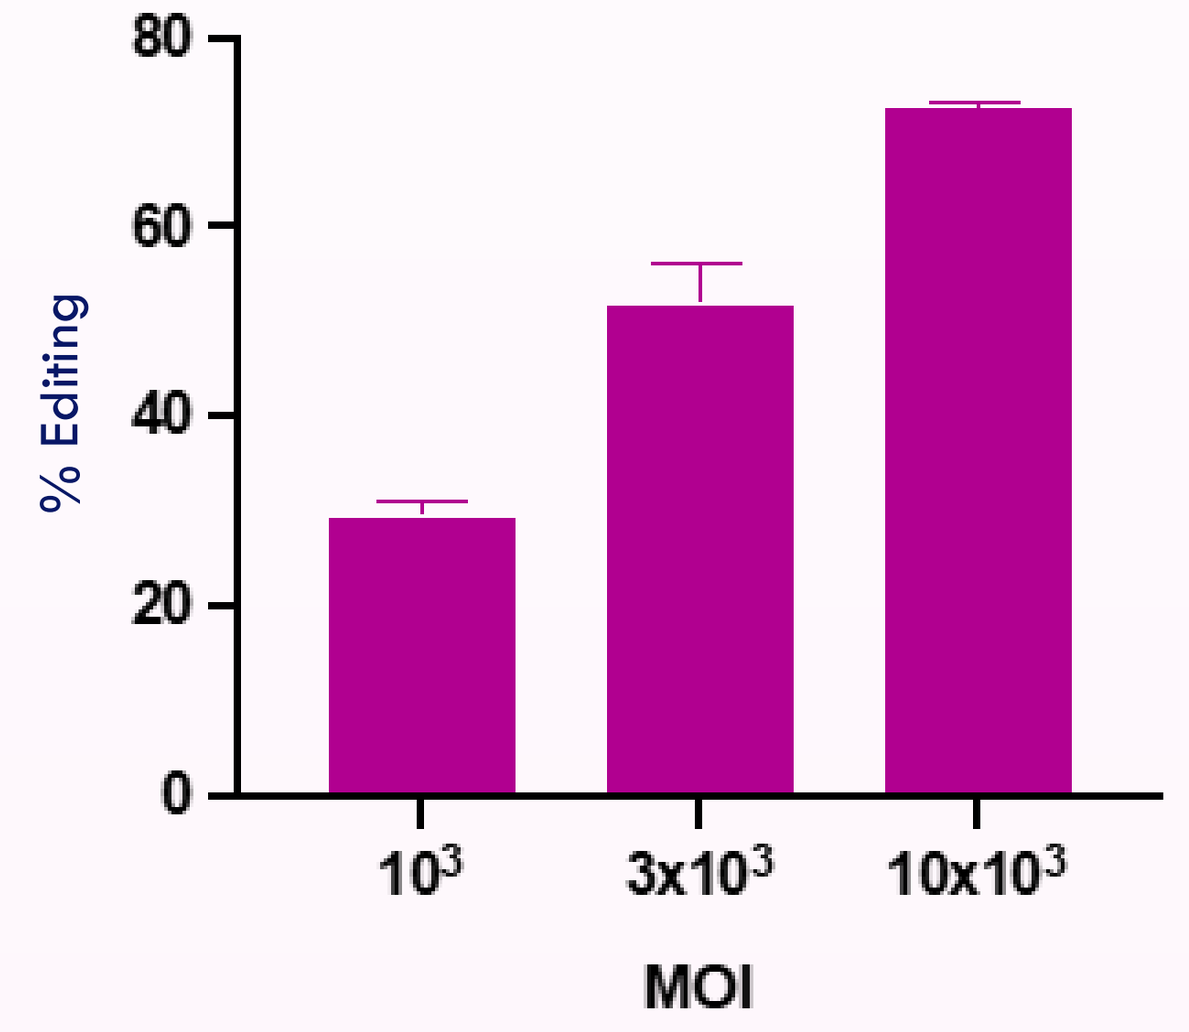

AAV mediated editing
% Editing
10^3 $3x10^3$ $10x10^3$
MOI

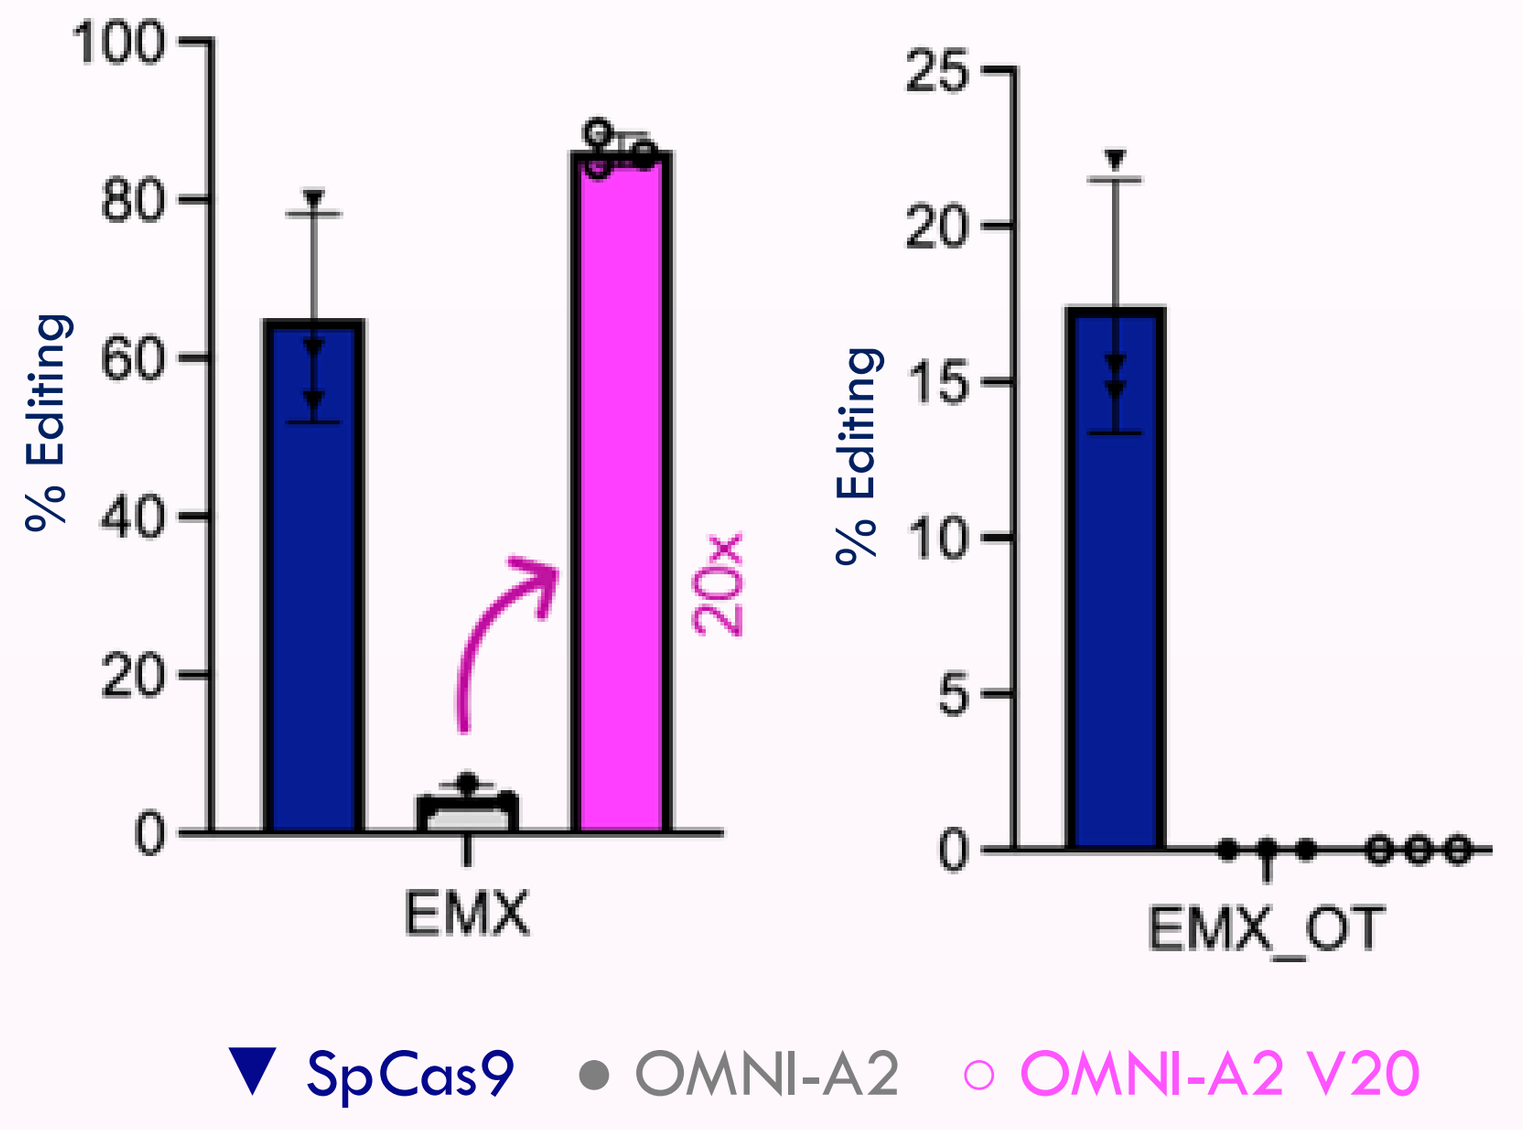

On targets
% Editing
20x
EMX
Off targets
% Editing
EMX_OT
▼ SpCas9 ● OMNI-A2 ○ OMNI-A2 V20

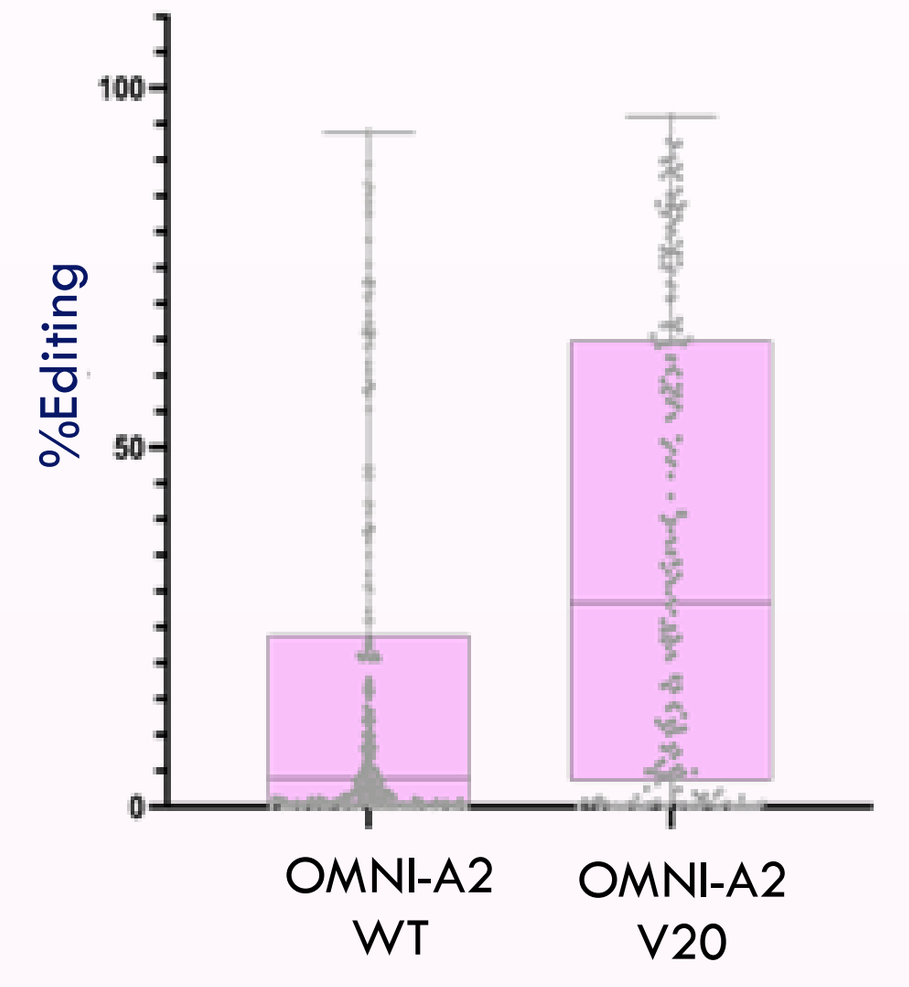

Improved editing of OMNI-A2 variant
%Editing
OMNI-A2 WT
OMNI-A2 V20

3. OMNI-A13- Applying AI-based engineering to improve protein stability

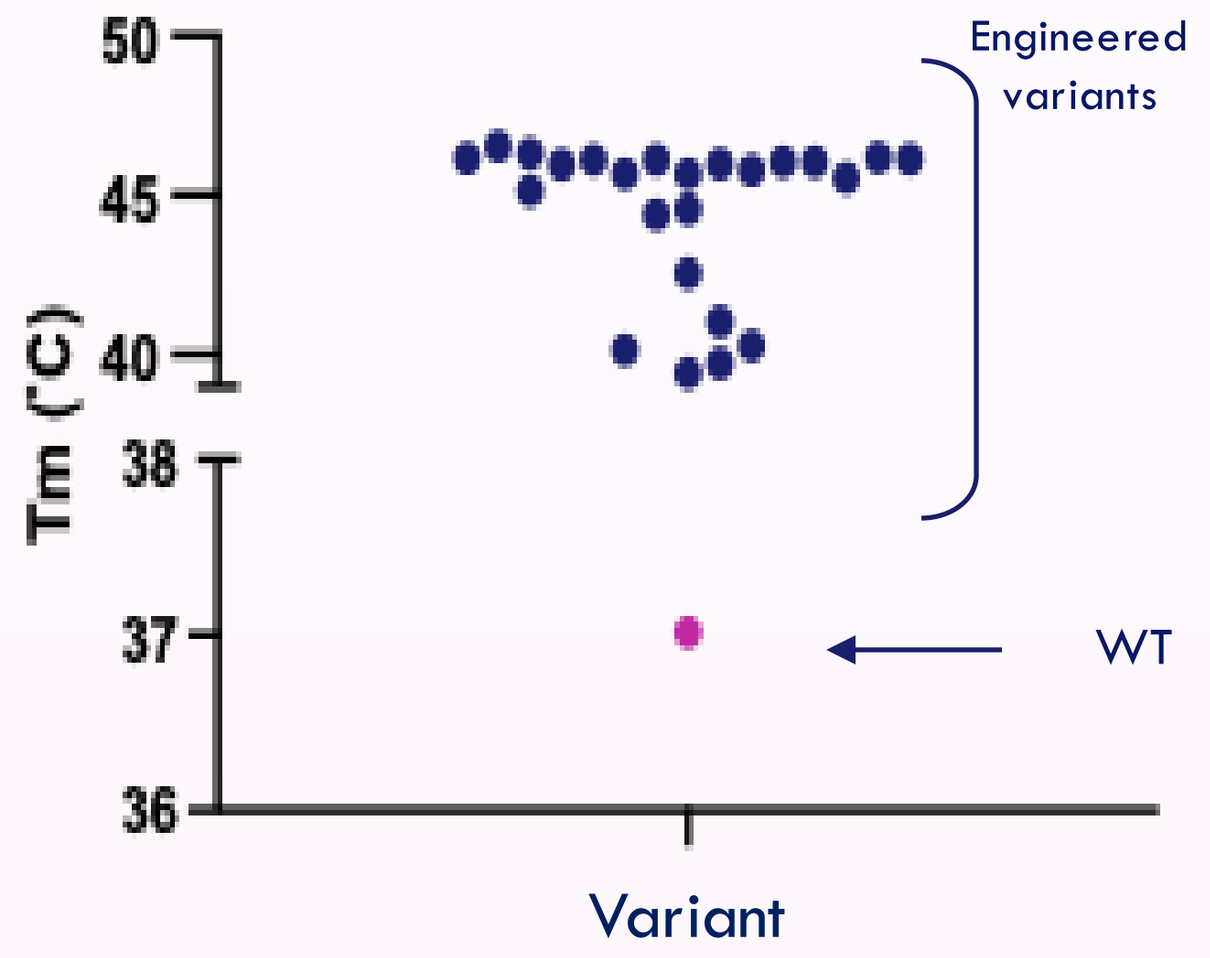

Thermal stability
Tm (°C)
Engineered variants
WT
Variant

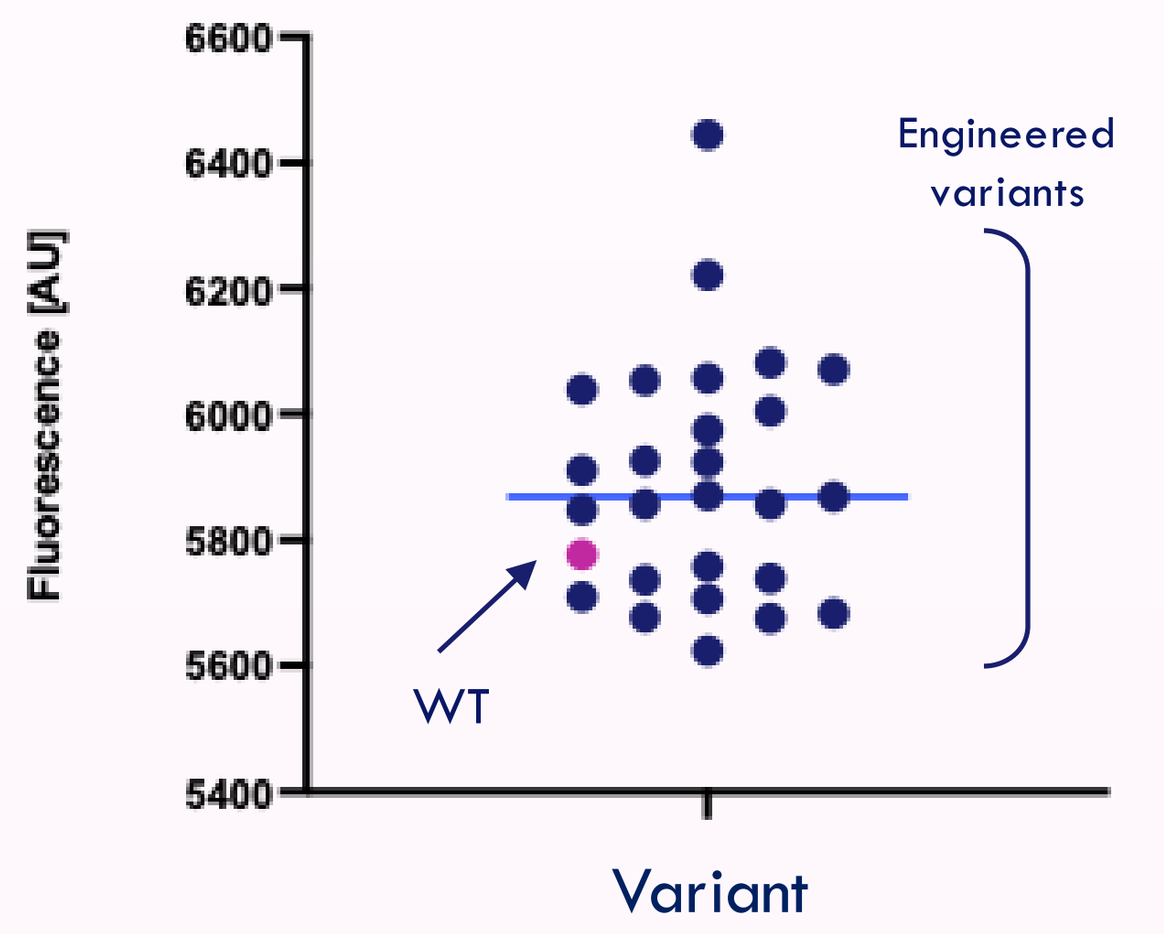

In-vitro activity
Fluorescence [AU]
Engineered variants
WT
Variant

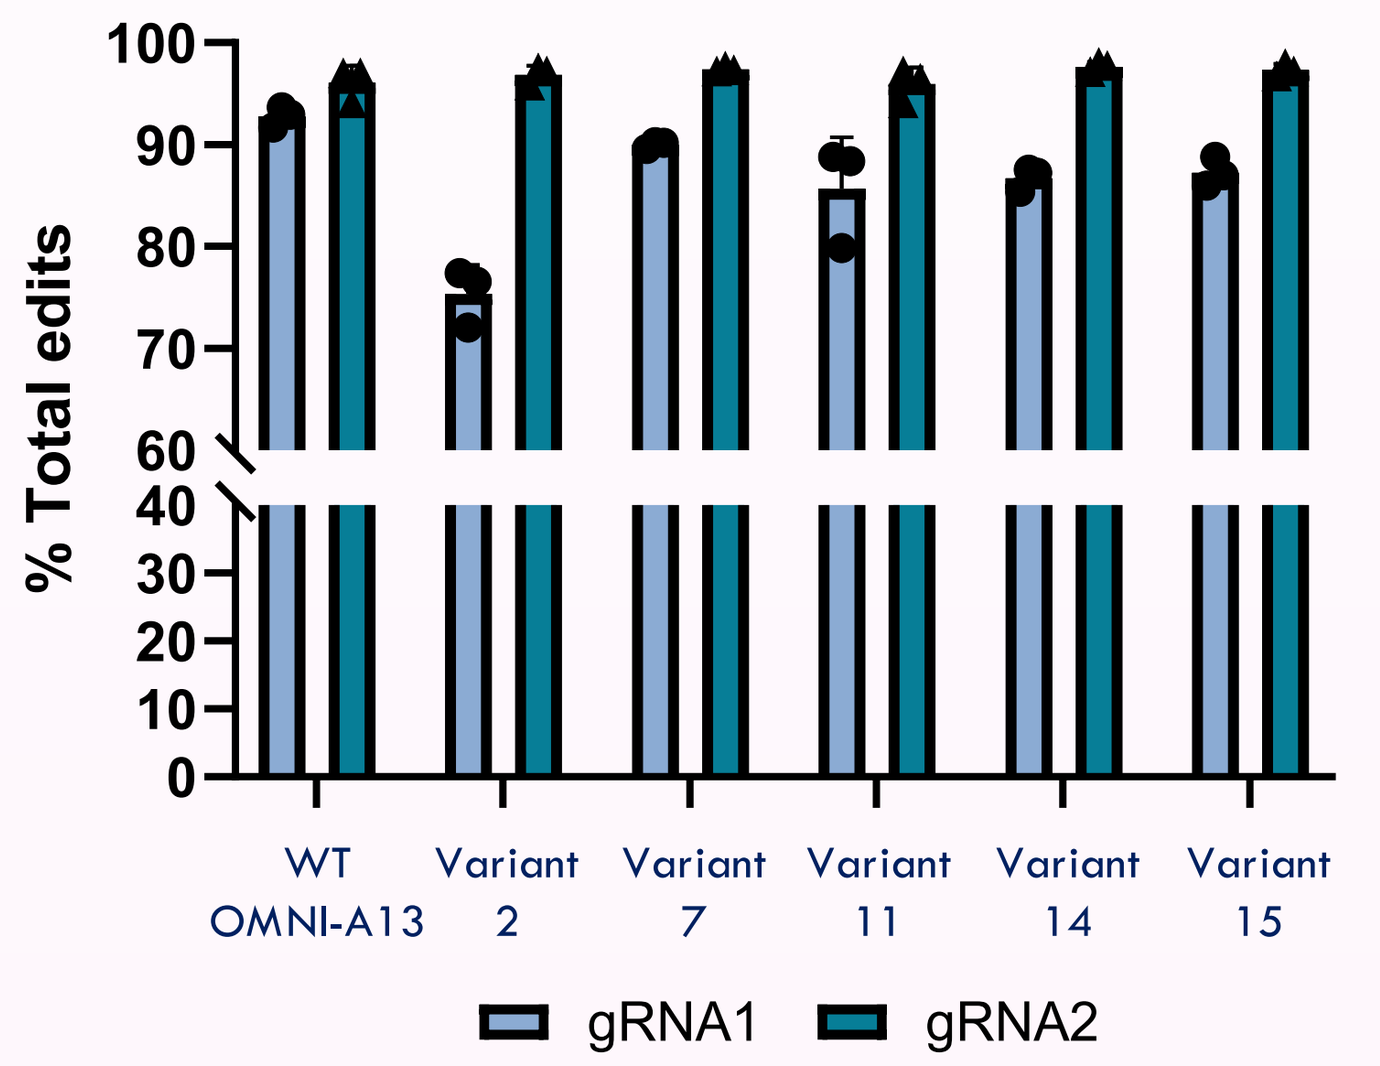

In-vivo activity
% Total edits
WT OMNI-A13
Variant 2
Variant 7
Variant 11
Variant 14
Variant 15
gRNA1 gRNA2

Non-NGG-PAM nuclease enables efficient editing to overcome IP constrains while maintaining efficiency and safety

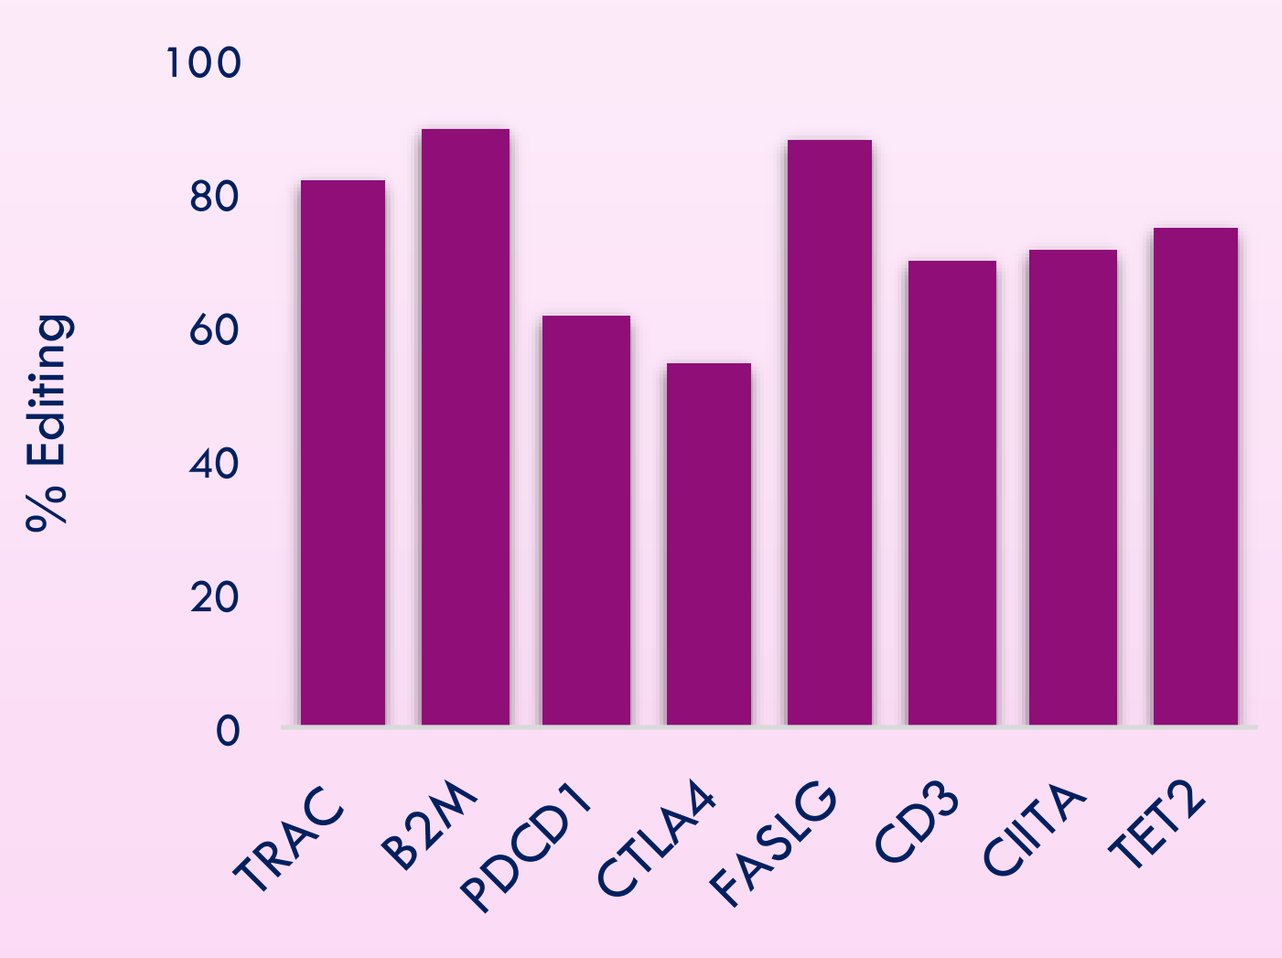

T Cells
% Editing
TRAC B2M PDCD1 CTLA4 FASLG CD3 CIITA TET2

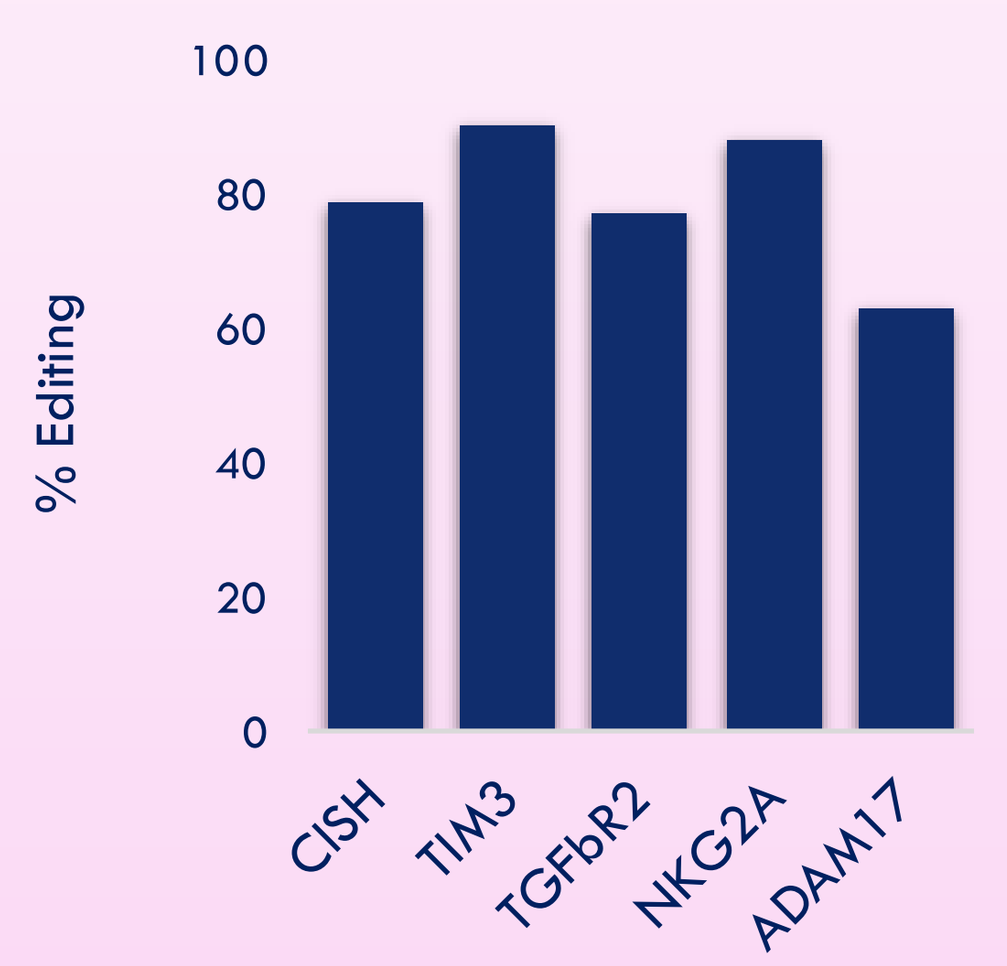

NK Cells
% Editing
CISH TIM3 TGFbR2 NKG2A ADAM17

Efficient protein knock-out

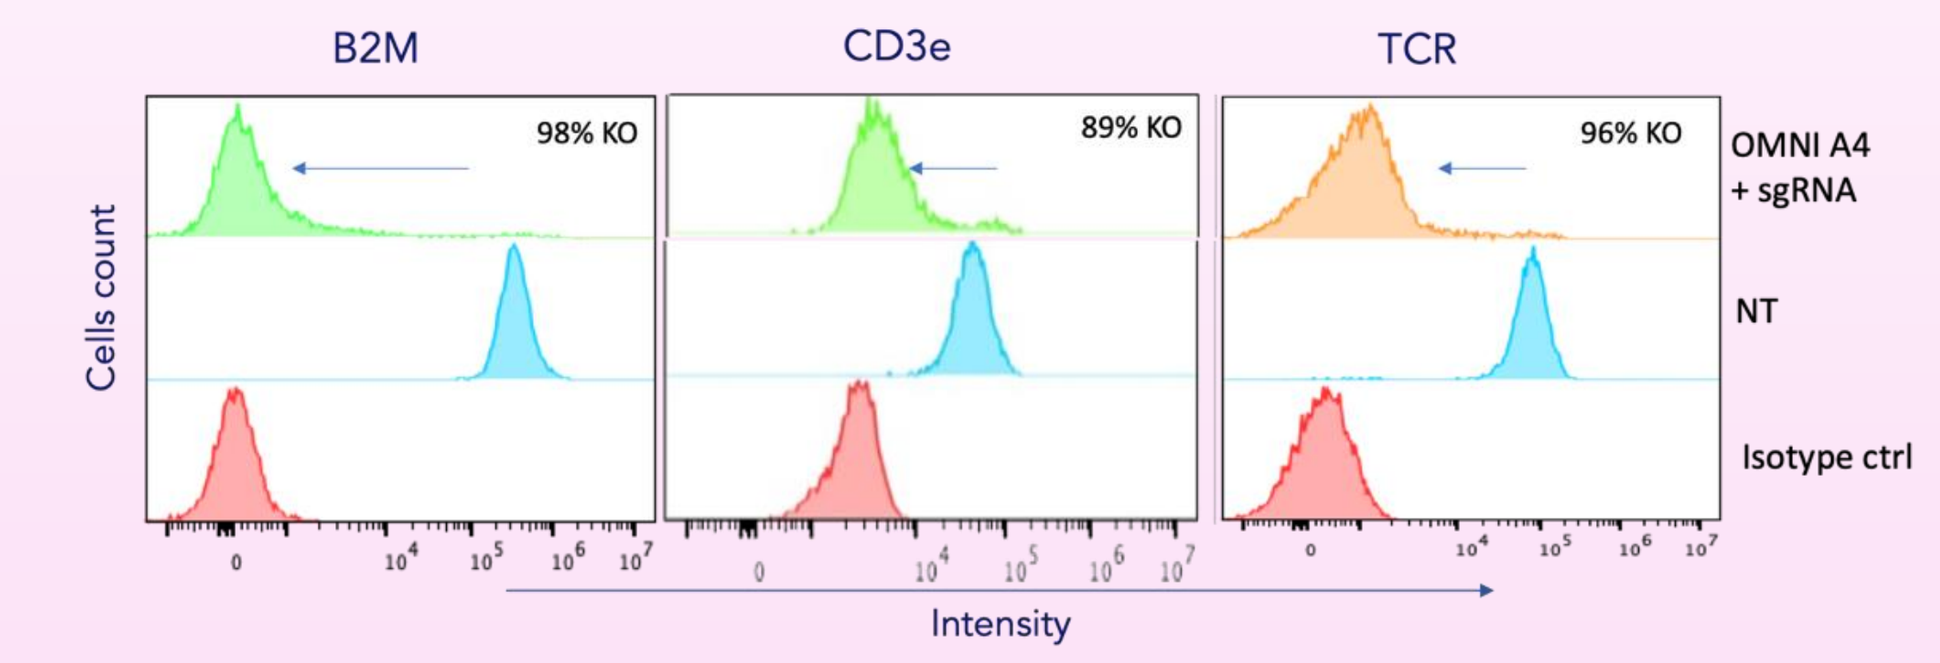

B2M
CD3e
TCR
98% KO
89% KO
96% KO
OMNI A4 + sgRNA
NT
Isotype ctrl
Cells count
Intensity

No off-target detected

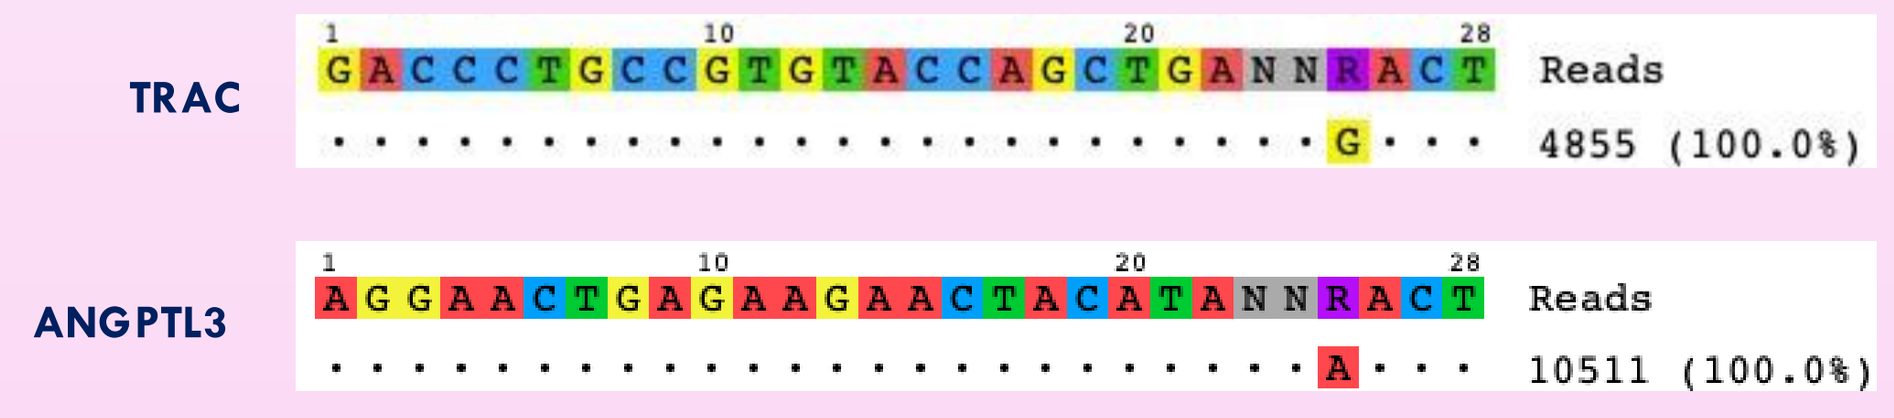

TRAC
Reads
4855 (100.0%)
ANGPTL3
Reads
10511 (100.0%)

Next-Generation OMNI-based tools for DNA writing and HDR

1. OMNI-editor- enables effective DNA writing

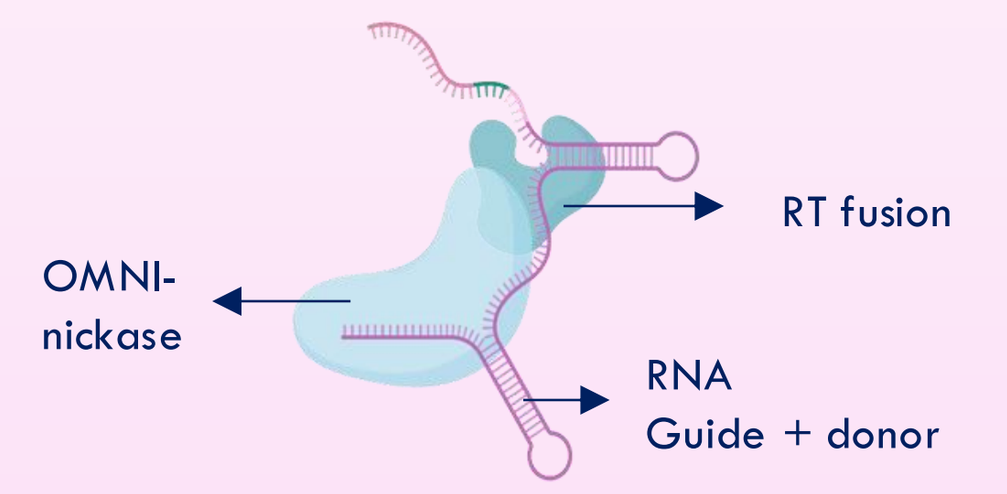

RT fusion
OMNI-nickase
RNA Guide + donor

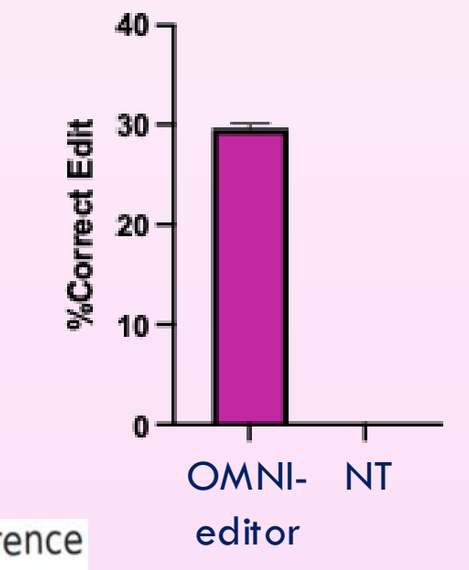

%Correct Edit
OMNI-editor
NT

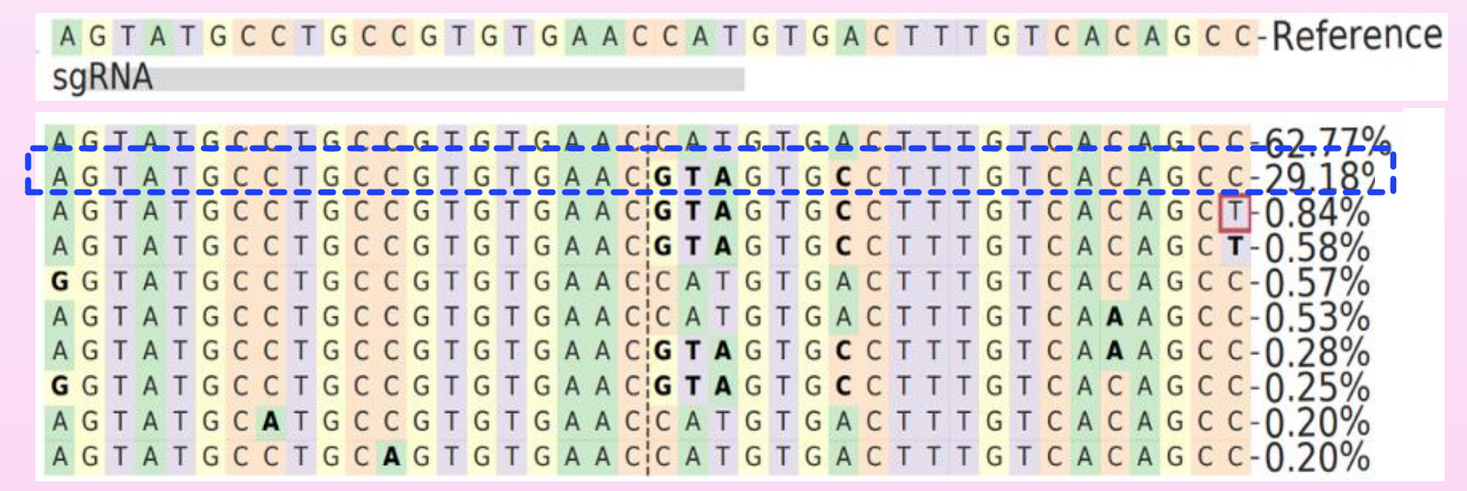

Reference
sgRNA
62.77%
29.18%
0.84%
0.58%
0.57%
0.53%
0.28%
0.25%
0.20%
0.20%

2. High HDR efficiency using OMNI-nucleases

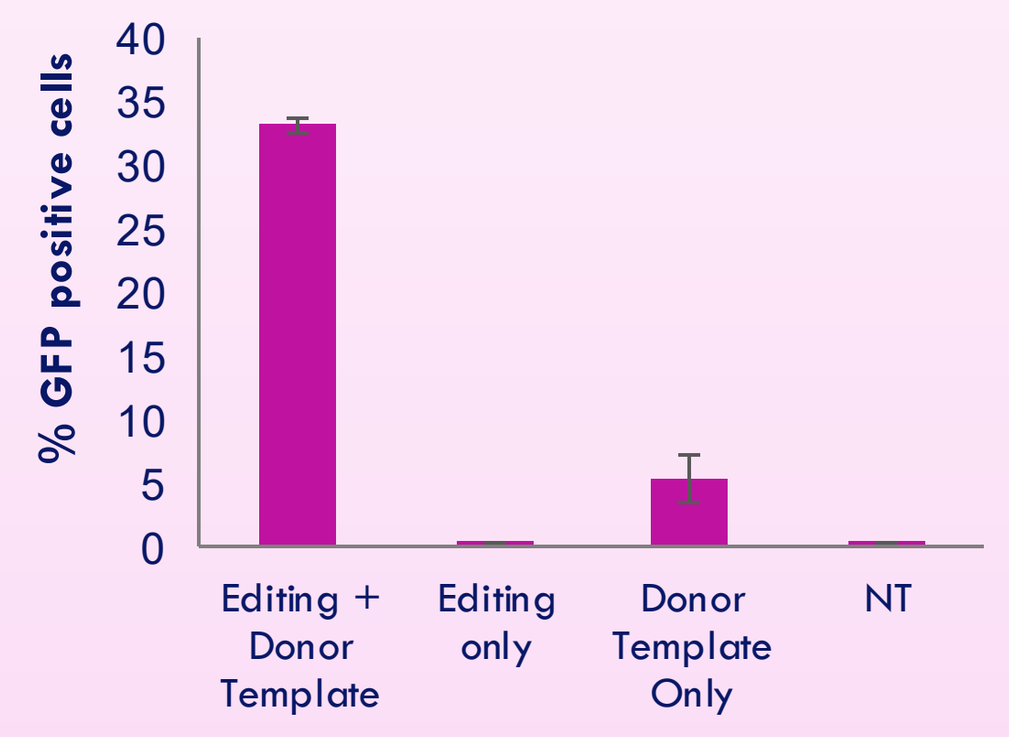

% GFP positive cells
Editing + Donor Template
Editing only
Donor Template Only
NT

Safe harbor site - locus 1
HepG2 cells

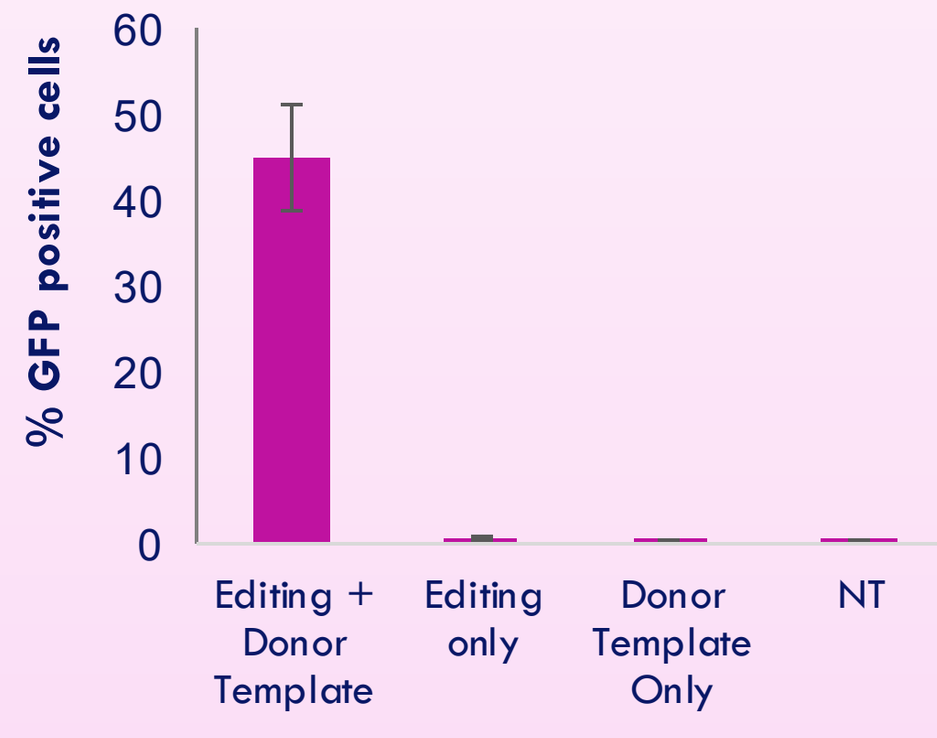

% GFP positive cells
Editing + Donor Template
Editing only
Donor Template Only
NT

Safe harbor site - locus 2
Primary HSCs

3. OMNI-off systems are in development

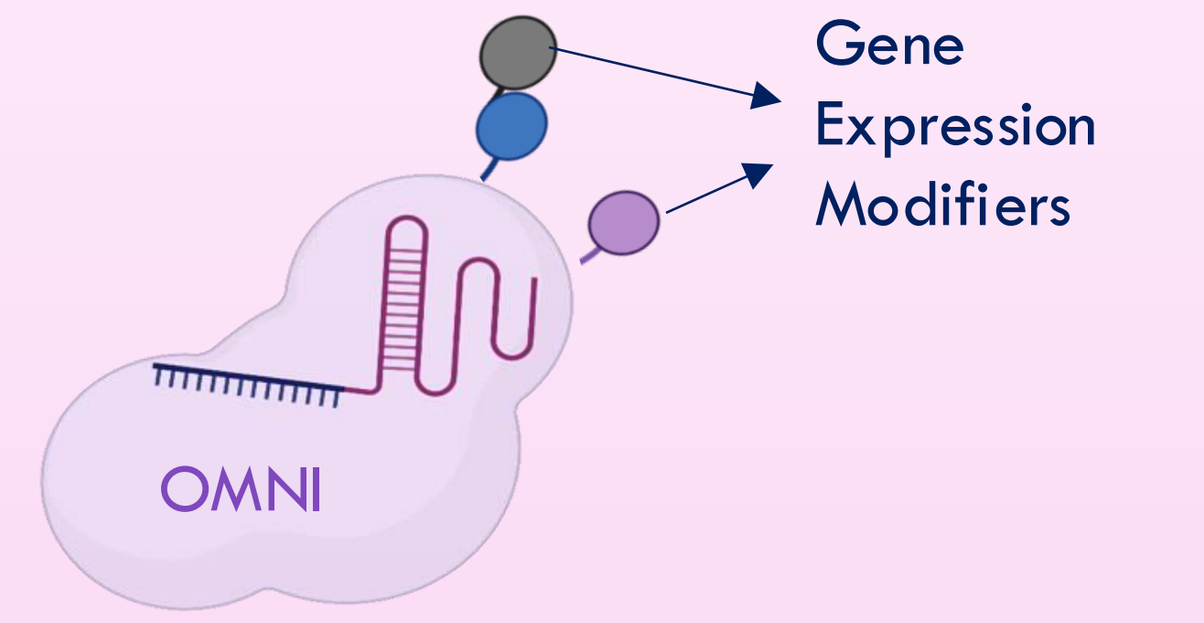

Gene Expression Modifiers
OMNI